Discovery and Activity of STRO-002, a Novel ADC Targeting Folate Receptor Alpha for Ovarian and Endometrial Cancer

Xiaofan Li, Cristina Abrahams, Sihong Zhou, Stellanie Krimm, Robert Henningsen, Heather Stephenson, Jeffrey Hanson, Mary Rose Masikat, Krishna Bajjuri, Tyler Heibeck, Cuong Tran, Gang Yin, James Zawada, Ganapathy Sarma, Joy Chen, Maureen Bruhns, Willy Solis, Alexander Steiner, Adam Galan, Toni Kline, Ryan Stafford, Alice Yam, Venita I. De Almeida, Mark Lupher, Jr., Trevor Hallam.
Sutro Biopharma, South San Francisco, CA, USA

Introduction

- Folate receptor alpha (FolRα) is a cell-surface protein overexpressed in ovarian and endometrial cancer.
- FolRα expression is highly restricted on normal tissues, making it a promising target for cancer therapy using antibody drug conjugates (ADCs).
- We have used a platform based on Sutro's proprietary XpressCF+™ cell-free expression system and site specific conjugation to design a novel, FolRα-targeting ADC, STRO-002.
- STRO-002 contains the anti-FolRα human IgG1 antibody SP8166 (H01) conjugated to a novel proprietary cleavable drug-linker (SC239) at specific sites Y180 and F404 on the antibody heavy chain.
- SP8166 (H01) was discovered and optimized using a Fab ribosome display selection and has four non-natural amino acid, p-azido-phenylalanine (pAMF) residues incorporated at positions Y180 and F404 on each heavy chain.
- SC239 is composed of a tubulin-targeting 3-aminophenyl hemiasterlin warhead, SC209, and a cleavable valine citrulline p-aminobenzyl carbamate linker functionalized with dibenzocyclooctyne (DBCO).
- The rapid and selective reaction of DBCO on SC239 and pAMF residues in SP8166 results in a well-defined, homogeneous ADC with a drug-antibody ratio (DAR) of ~4.

Generation of STRO-002, A Homogenous FolRα-Targeting ADC Through Cell-Free Antibody Synthesis And Site-Specific Conjugation

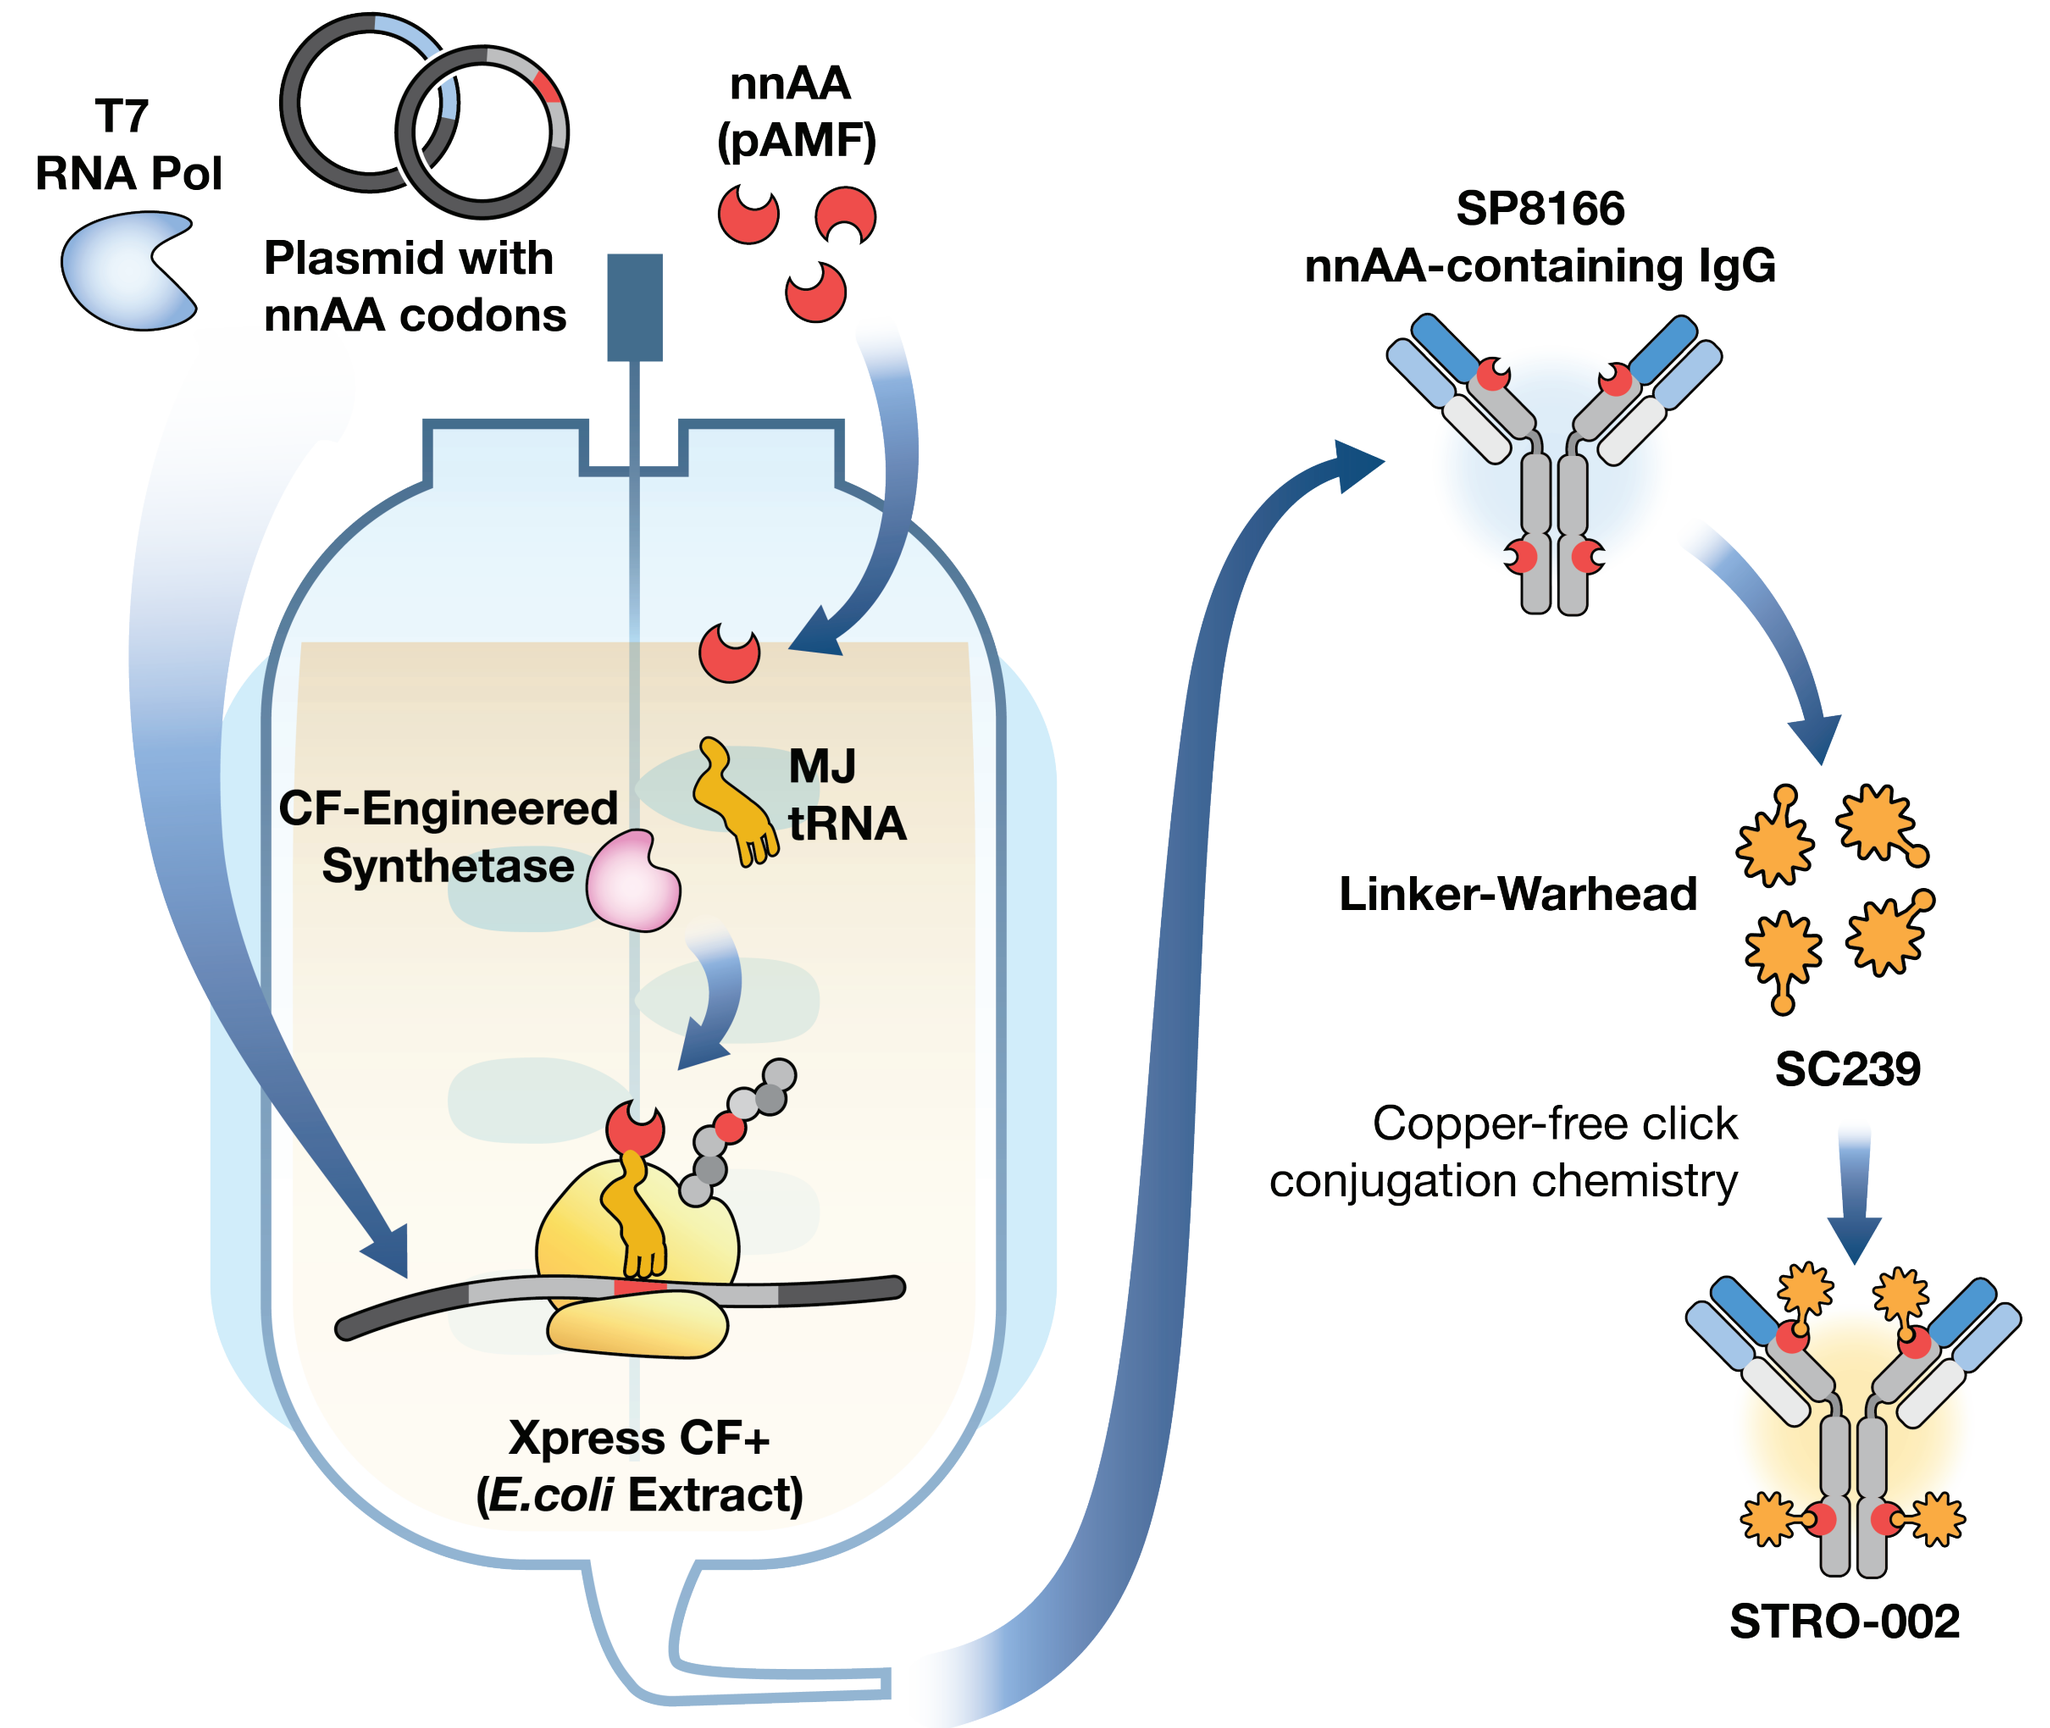
T7
RNA Pol
Plasmid with
nnAA codons
nnAA
(pAMF)
CF-Engineered
Synthetase
MJ
tRNA
Xpress CF+
(E.coli Extract)
SP8166
nnAA-containing IgG
Linker-Warhead
SC239
Copper-free click
conjugation chemistry
STRO-002

Sutro's proprietary XpressCF+™ cell-free expression system includes:

- incorporation of pAMF, that enables site-specific conjugation,
- an orthogonal tRNA synthetase which enables high fidelity pAMF incorporation,
- engineered XpressCF+™ extracts that utilize an engineered, attenuated RF-1 which enables:
 - a. Efficient and multiple insertion points of pAMF in the same translation product
 - b. Precise DAR ranging from 1-8 in a single molecular species ADC
 - c. ADC production in a few days allowing for rapid iterative structure-activity optimization
- anti-FolRα antibodies isolated using Fab-based ribosome display were optimized for
 - Linker and warhead attributes
 - Antibody moiety
 - Drug antibody ratio
 - Site pairs for drug-linker attachment

Selection of Optimum Drug-Linker and DAR
ADC with a Cleavable Drug-Linker at DAR 4 Showed Best Activity

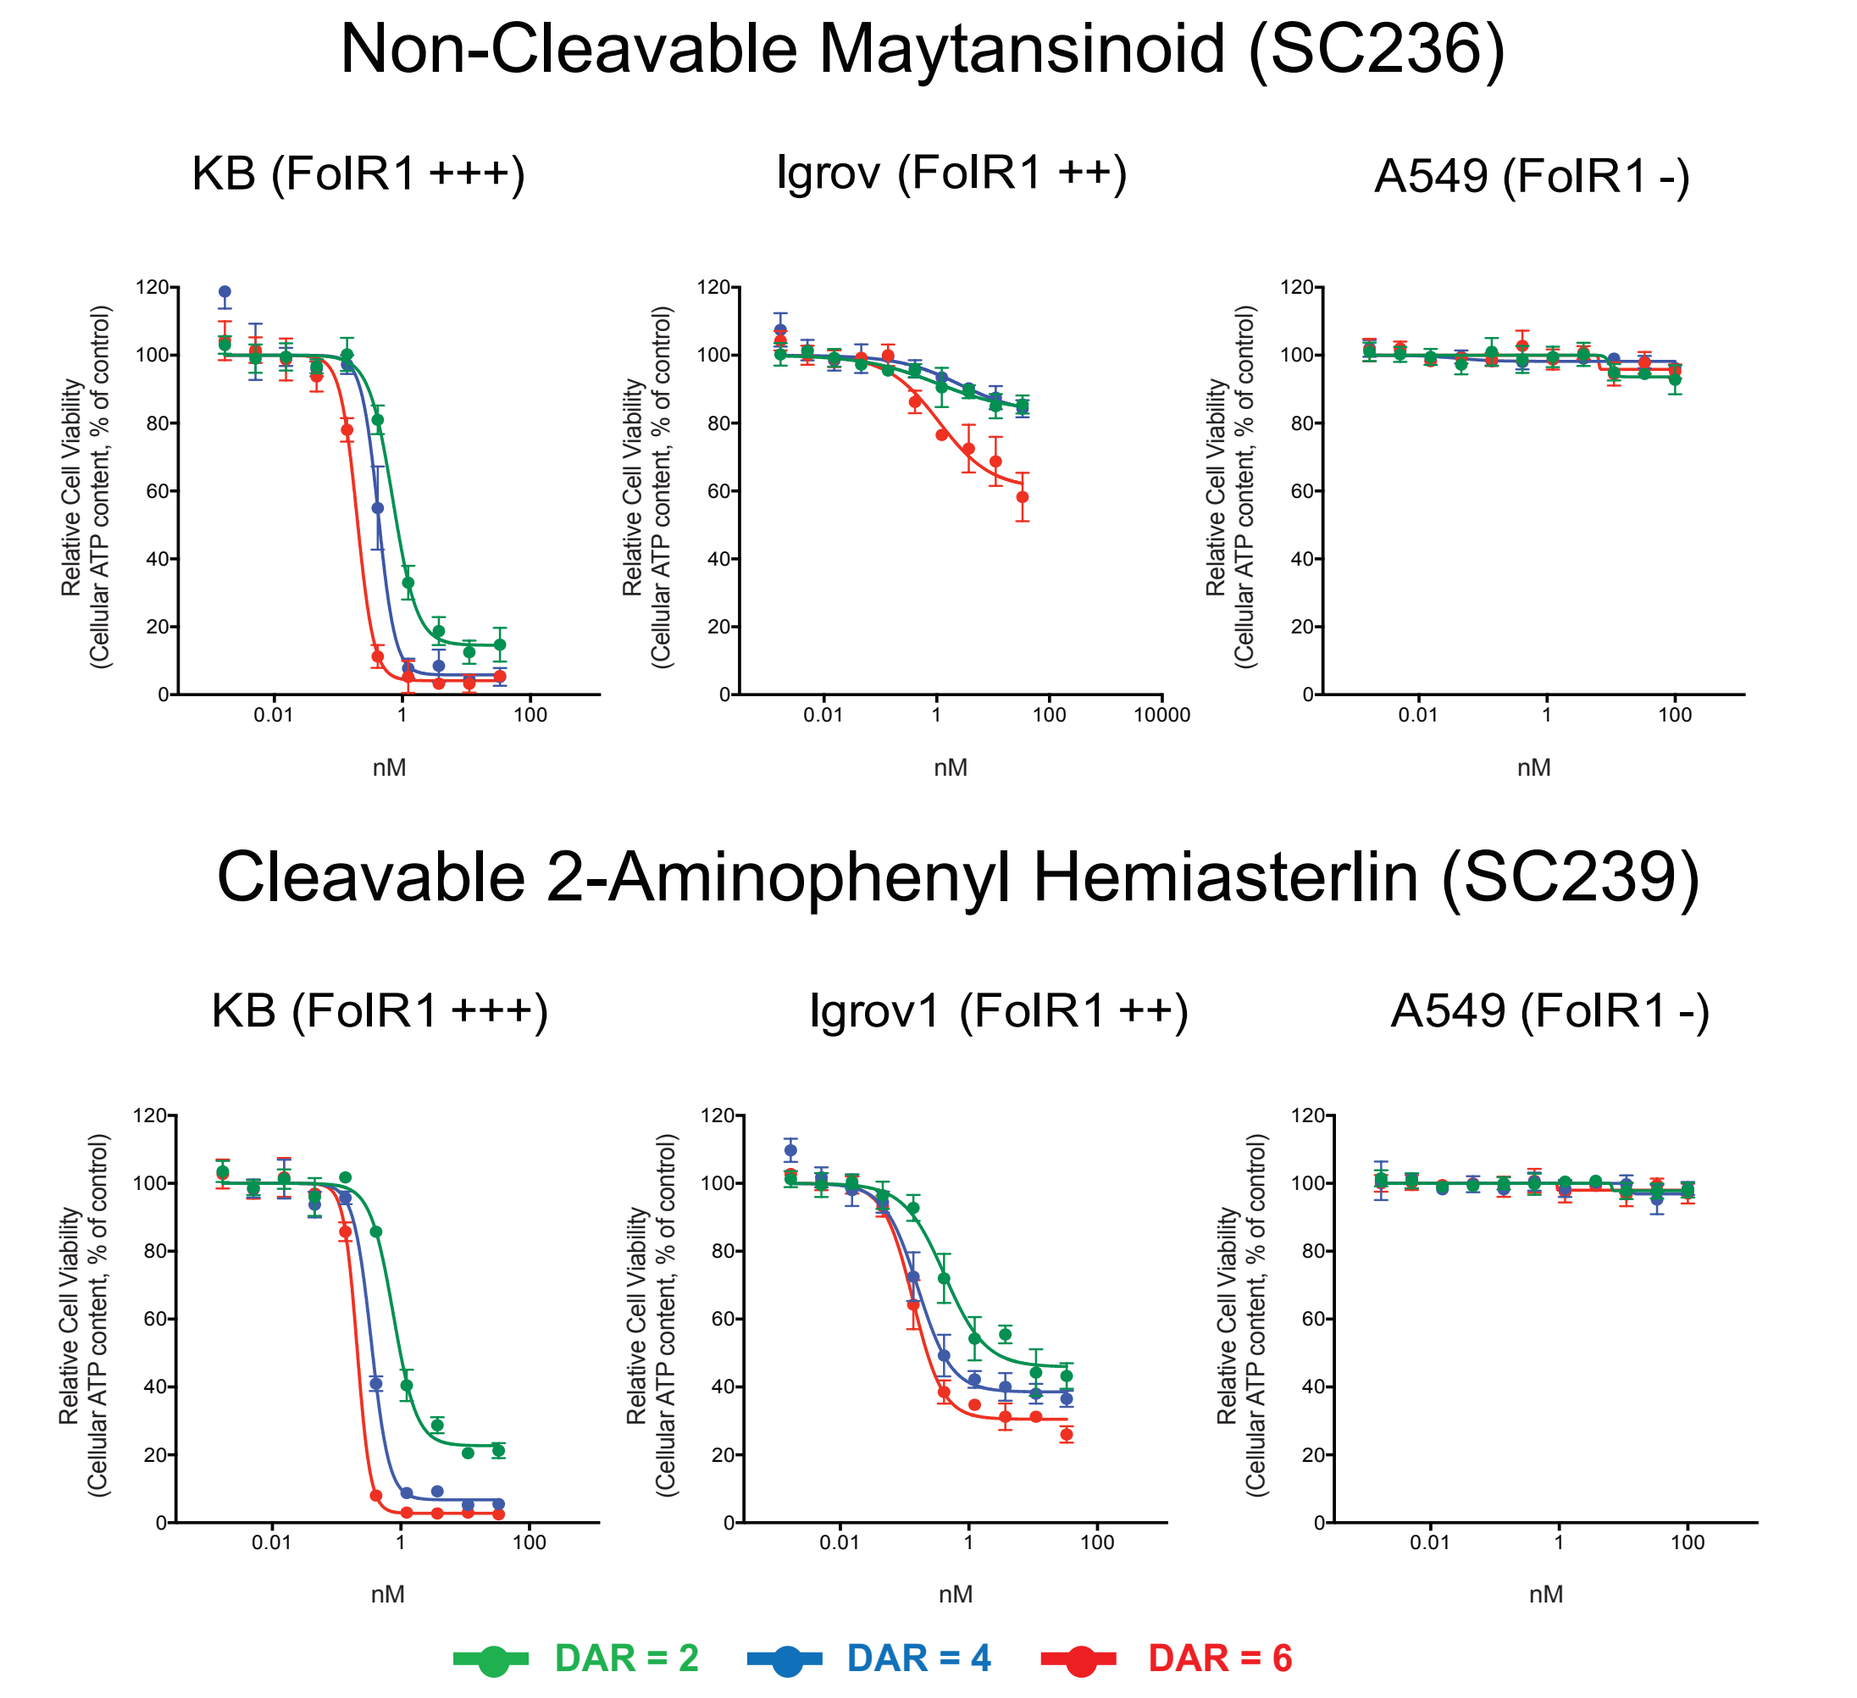
Non-Cleavable Maytansinoid (SC236)
KB (FolR1 +++)
Igrov (FolR1 ++)
A549 (FolR1 -)
Cleavable 2-Aminophenyl Hemiasterlin (SC239)
KB (FolR1 +++)
Igrov1 (FolR1 ++)
A549 (FolR1 -)
Relative Cell Viability (Cellular ATP content, % of control)
nM
DAR = 2
DAR = 4
DAR = 6

- FolRα ADC variants with different tubulin based drug-linkers, a non-cleavable maytansinoid drug-linker (SC236) or a cleavable 2-aminophenyl hemiasterlin drug-linker (SC239), conjugated to FolRα B10 antibody at DAR = 2, 4, or 6, using the Y180, F404 and/or K42 sites were compared.
- SC239 conjugates exhibited better cytotoxic activity on Igrov1 cells which have lower, but clinically relevant expression levels of FolRα.
- SC239 was therefore selected as the drug-linker for this ADC.
- A conjugate with a DAR of 4 was selected since a DAR of 6 did not provide much higher cytotoxicity than the DAR 4 version.

Selection of Top Antibodies
H01 and B10 FolRα Antibodies Exhibited Best Activity in FolRα Expressing Models

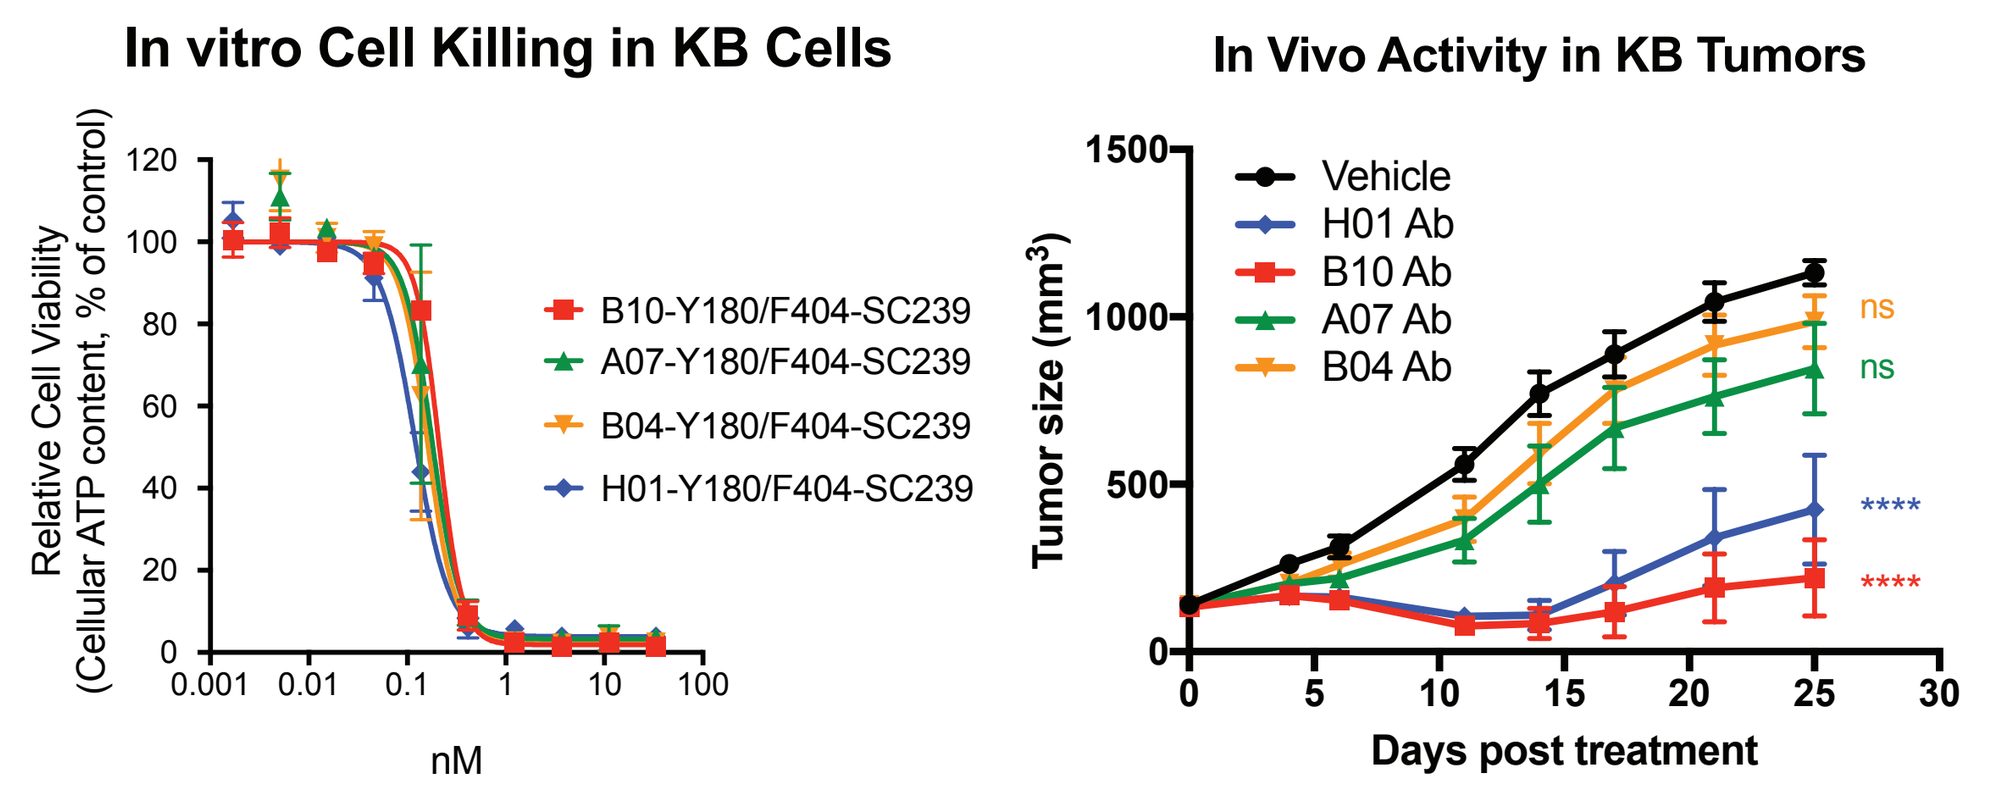
In vitro Cell Killing in KB Cells
Relative Cell Viability (Cellular ATP content, % of control)
B10-Y180/F404-SC239
A07-Y180/F404-SC239
B04-Y180/F404-SC239
H01-Y180/F404-SC239
nM
In Vivo Activity in KB Tumors
Tumor size (mm³)
Vehicle
H01 Ab
B10 Ab
A07 Ab
B04 Ab
ns
ns

Days post treatment

- FolRα ADC variants with different FolRα antibodies (engineered and isolated from Fab ribosome display) conjugated to SC239 at positions Y180 and F404 were compared.
- All variants had comparable *in vitro* cell killing on high expressing FolRα positive KB cells.
- In response to a single 2.5 mg/kg dose, ADC variants with H01 and B10 FolRα antibodies showed significant *in vivo* activity in the KB xenograft model.

Selection of Optimum Conjugation Sites
Conjugation at Sites Y180/F404 or Y180/K42 Showed Best *In Vivo* Activity

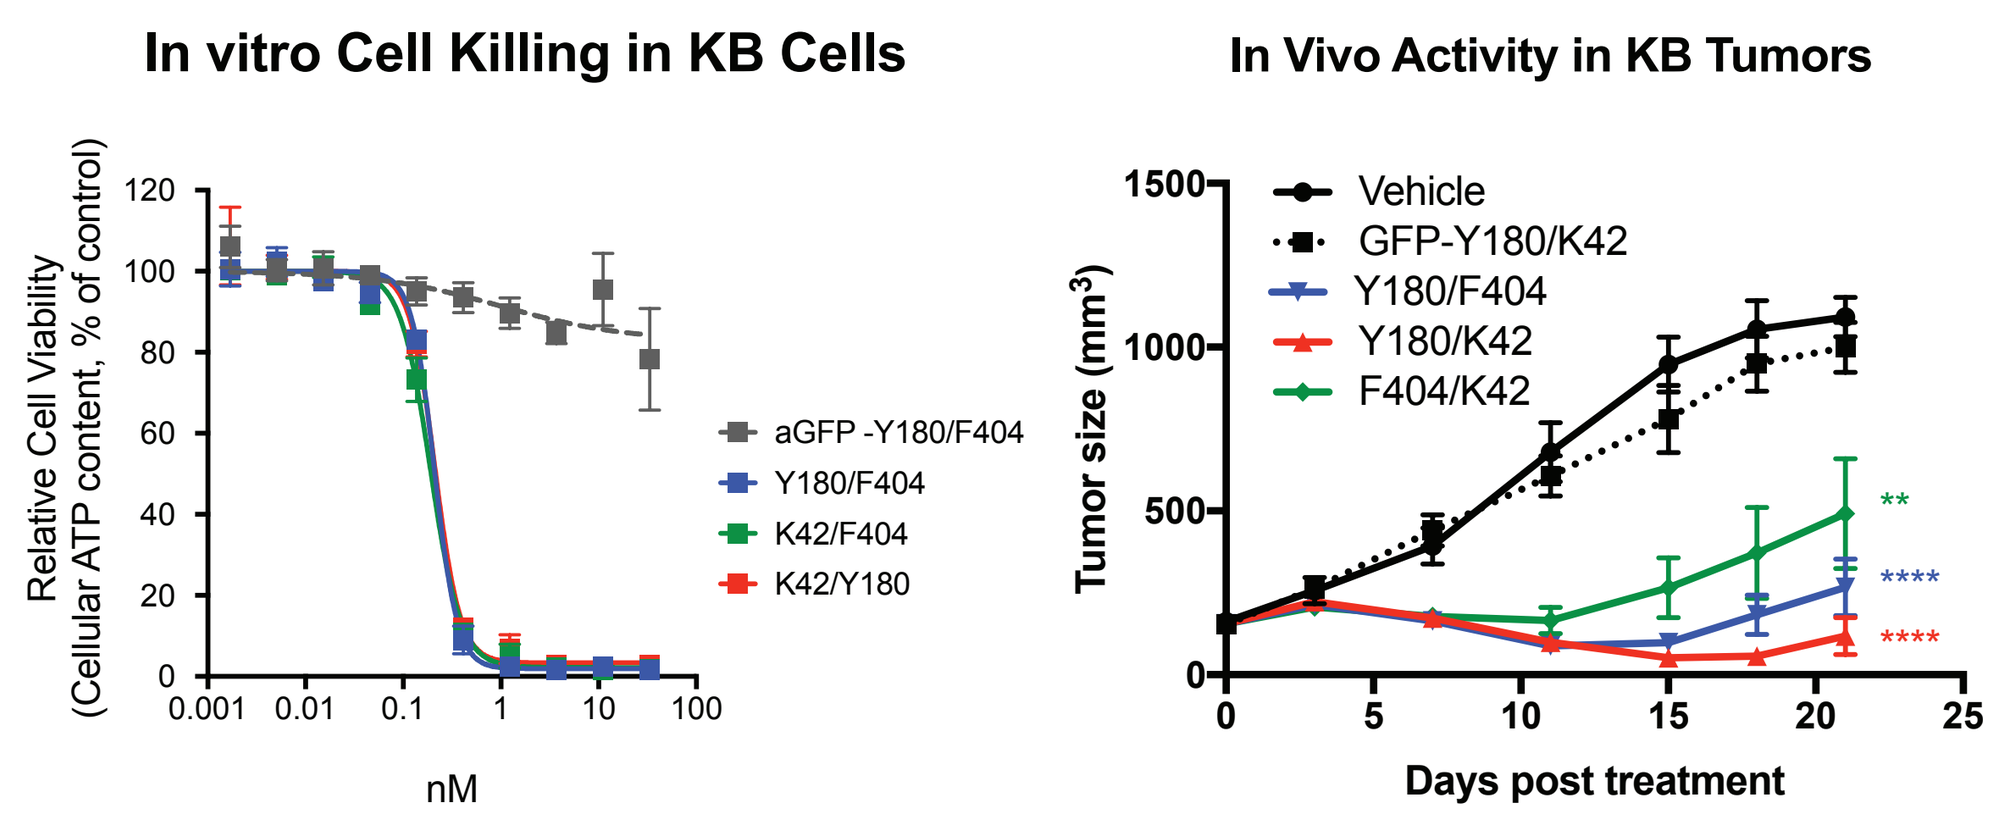
In vitro Cell Killing in KB Cells
Relative Cell Viability (Cellular ATP content, % of control)
aGFP -Y180/F404
Y180/F404
K42/F404
K42/Y180
nM
In Vivo Activity in KB Tumors
Tumor size (mm³)
Vehicle
GFP-Y180/K42
Y180/F404
Y180/K42
F404/K42
**

Days post treatment

- FolRα ADC variants composed of B10 FolRα antibody conjugated to SC239 at the following site pairs Y180/K42, Y180/F404 or F404/K42 were compared.
- All variants had comparable in vitro cell killing activity on KB cells.
- In response to a single dose at 2.5 mg/kg, ADC variants with SC239 conjugated at Y180/K42 or Y180/F404 demonstrated better *in vivo* activity in the KB xenograft model than the ADC conjugated at F404/K42.

Selection of Final ADC Format
FolRα antibody H01 Conjugated at Y180 and F404 Induced Significant Growth Inhibition of Igrov-1 Tumors

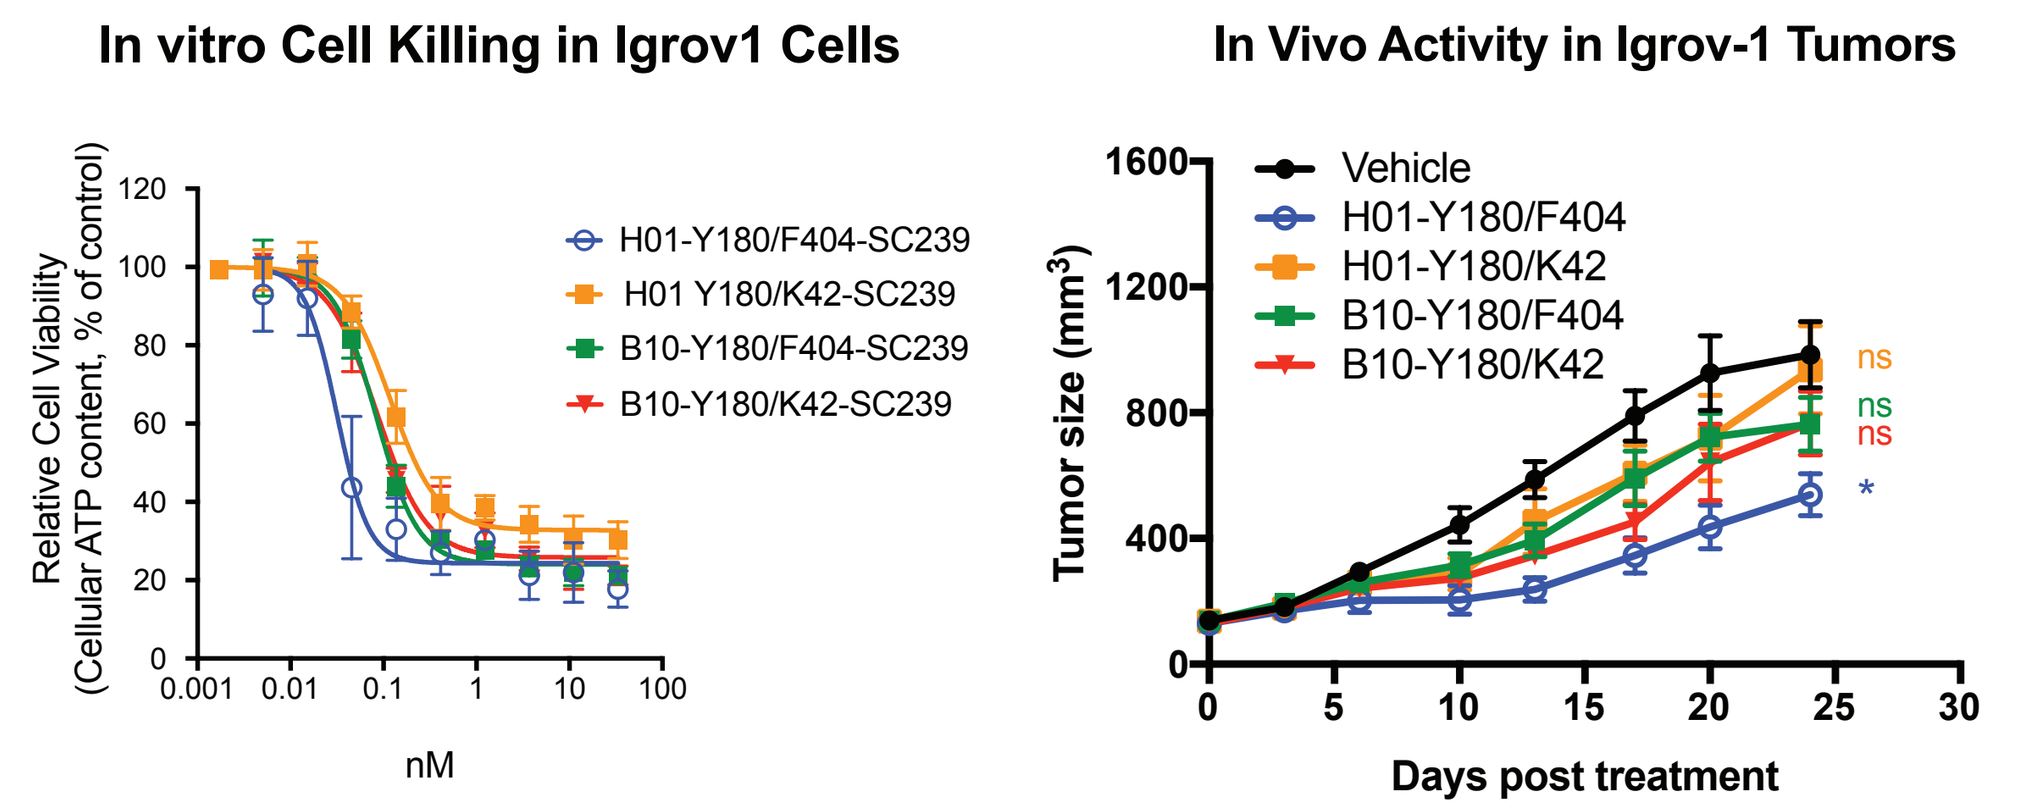
In vitro Cell Killing in Igrov1 Cells
Relative Cell Viability (Cellular ATP content, % of control)
H01-Y180/F404-SC239
H01 Y180/K42-SC239
B10-Y180/F404-SC239
B10-Y180/K42-SC239
nM
In Vivo Activity in Igrov-1 Tumors
Tumor size (mm³)
Vehicle
H01-Y180/F404
H01-Y180/K42
B10-Y180/F404
B10-Y180/K42
ns
ns
ns
*
Days post treatment

- FolRα ADC variants composed of B10 or H01 FolRα antibodies conjugated to SC239 linker warhead at Y180/K42 or Y180/F404 were compared.
- H01-Y180/F404 showed best *in vitro* cytotoxic activity and *in vivo* tumor growth inhibition in the Igrov-1 model following a single dose at 2.5 mg/kg.

Selection of Optimal Linker
Val-Cit Linker Confered Better Efficacy Than a Val-Ala Linker

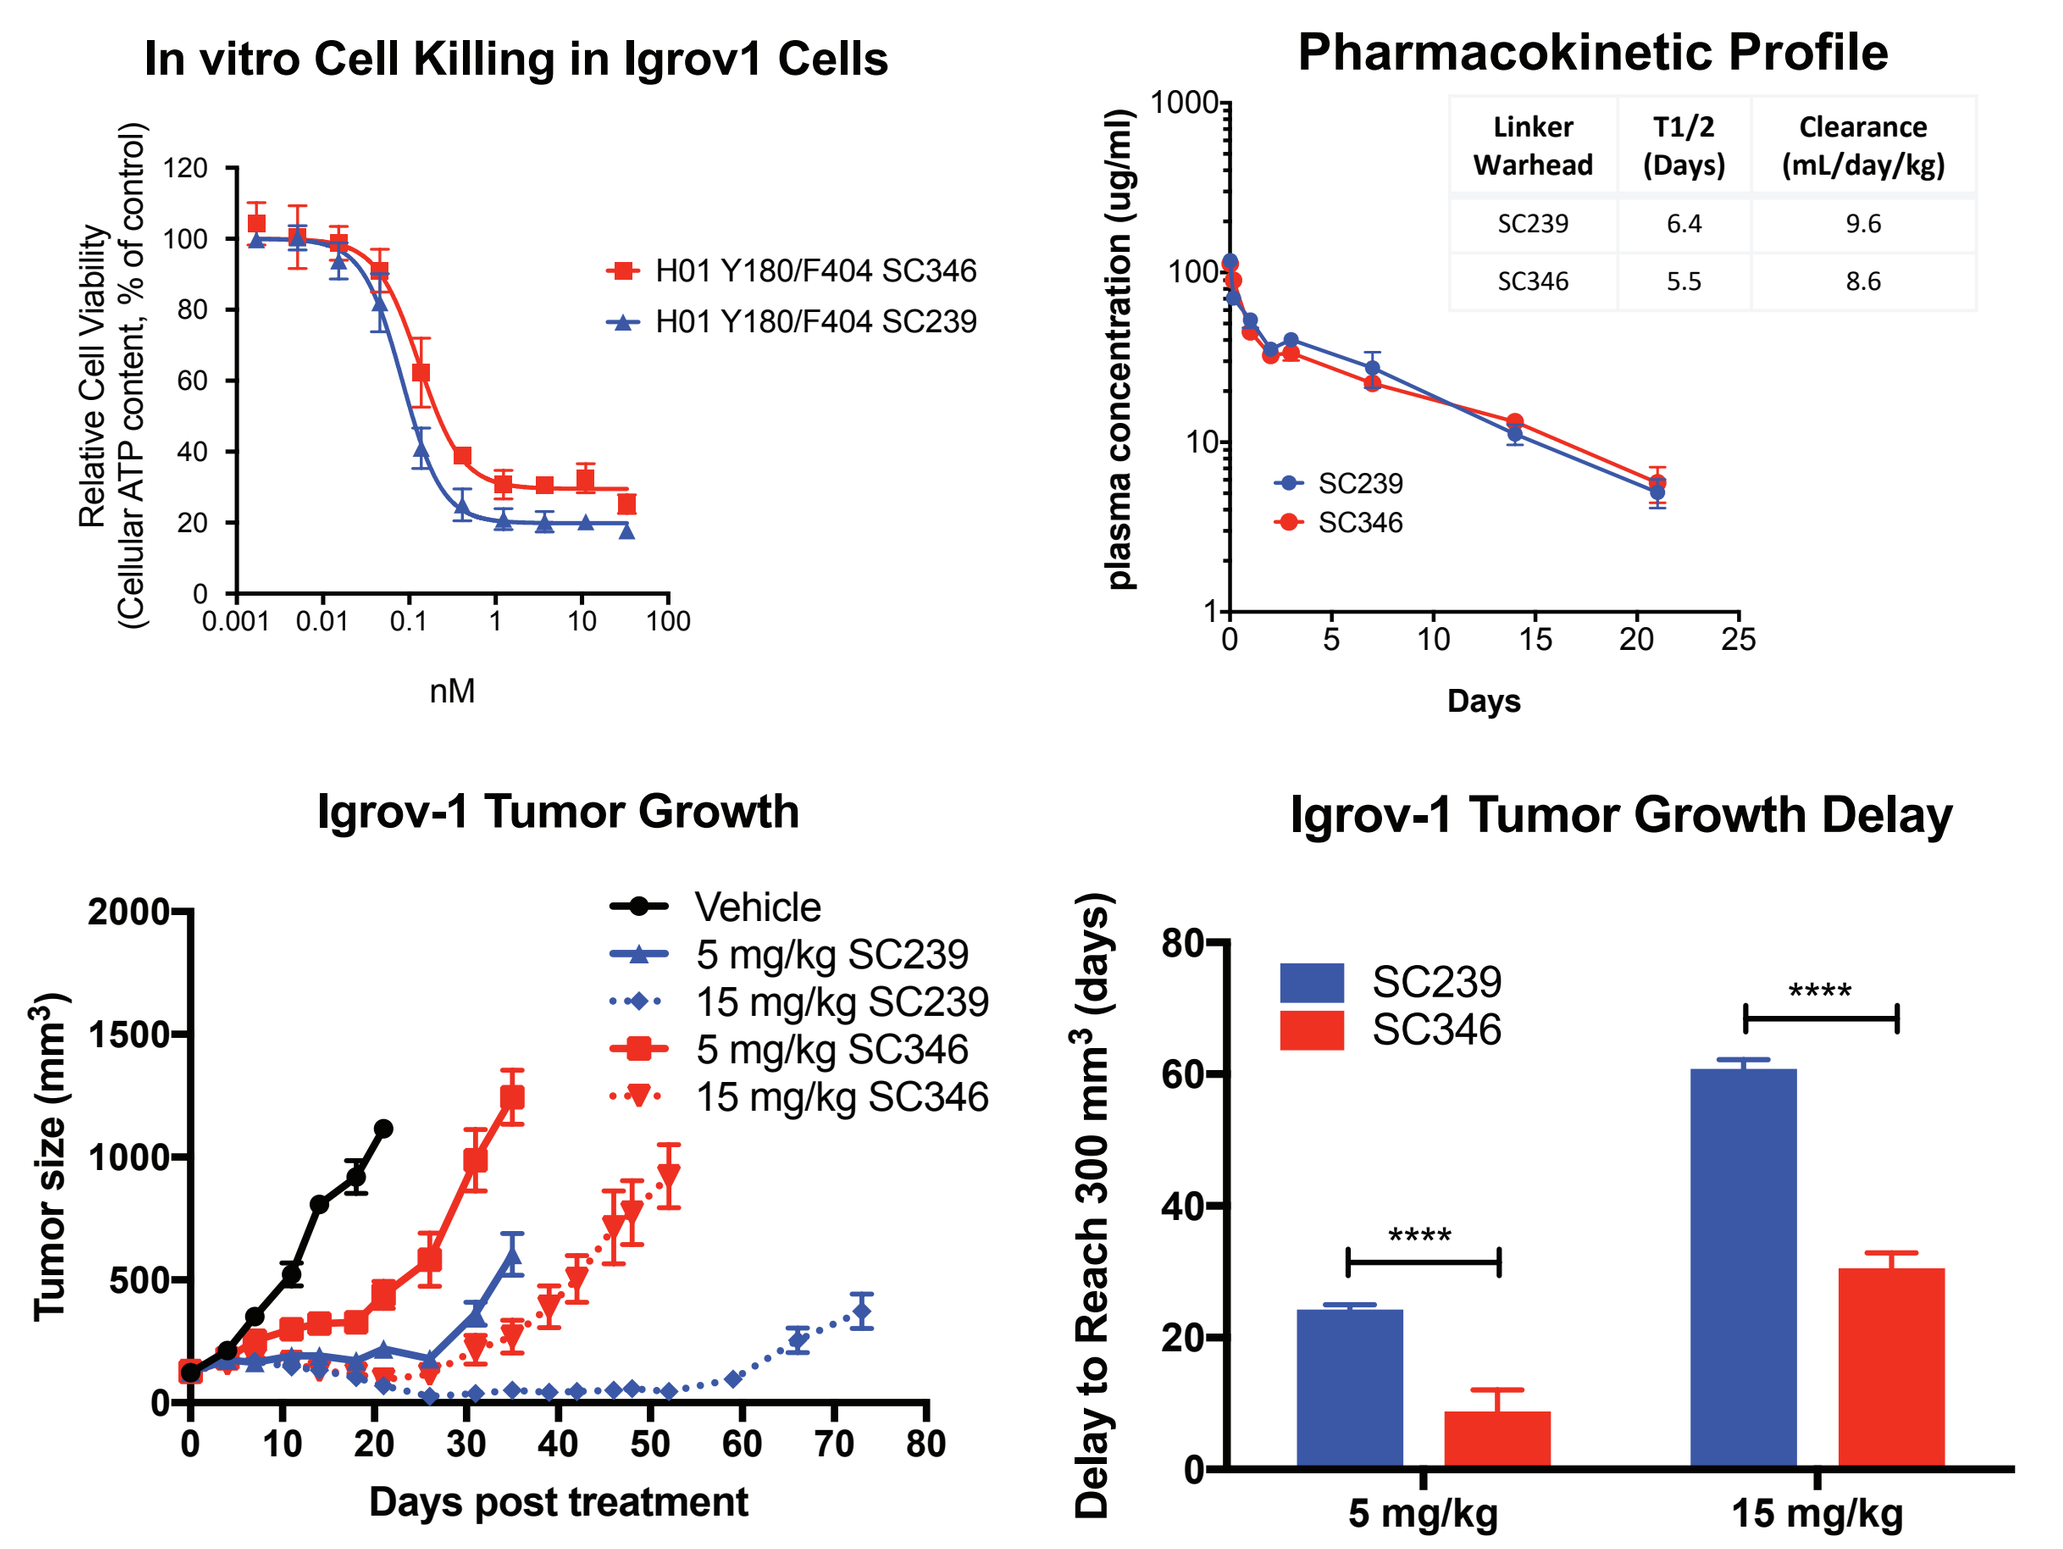
In vitro Cell Killing in Igrov1 Cells
Relative Cell Viability (Cellular ATP content, % of control)
H01 Y180/F404 SC346
H01 Y180/F404 SC239
nM
Pharmacokinetic Profile
Linker Warhead | T1/2 (Days) | Clearance (mL/day/kg)
SC239 | 6.4 | 9.6
SC346 | 5.5 | 8.6
plasma concentration (ug/ml)
SC239
SC346
Days
Igrov-1 Tumor Growth
Tumor size (mm³)
Vehicle
5 mg/kg SC239
15 mg/kg SC239
5 mg/kg SC346
15 mg/kg SC346
Days post treatment
Igrov-1 Tumor Growth Delay
Delay to Reach 300 mm³ (days)
SC239
SC346

5 mg/kg
15 mg/kg

- FolRα ADC variants composed of the H01-Y180/F404 FolRα antibody conjugated to 3-aminophenyl hemiasterlin with either a cleavable Val-Cit drug linker, or a Val-Ala drug linker, were compared.
- Both ADC variants showed comparable pharmacokinetic properties in mice.
- The SC239 conjugate showed slightly improved in vitro cell killing activity and exhibited superior in vivo efficacy than the SC346 conjugate in Igrov-1 model at a single dose of 5 and 15 mg/kg.
- Based on the cumulative data, H01 conjugated to SC239 at positions Y180 and F404 was selected as the lead FolRα targeting ADC, STRO-002..

Structure of STRO-002

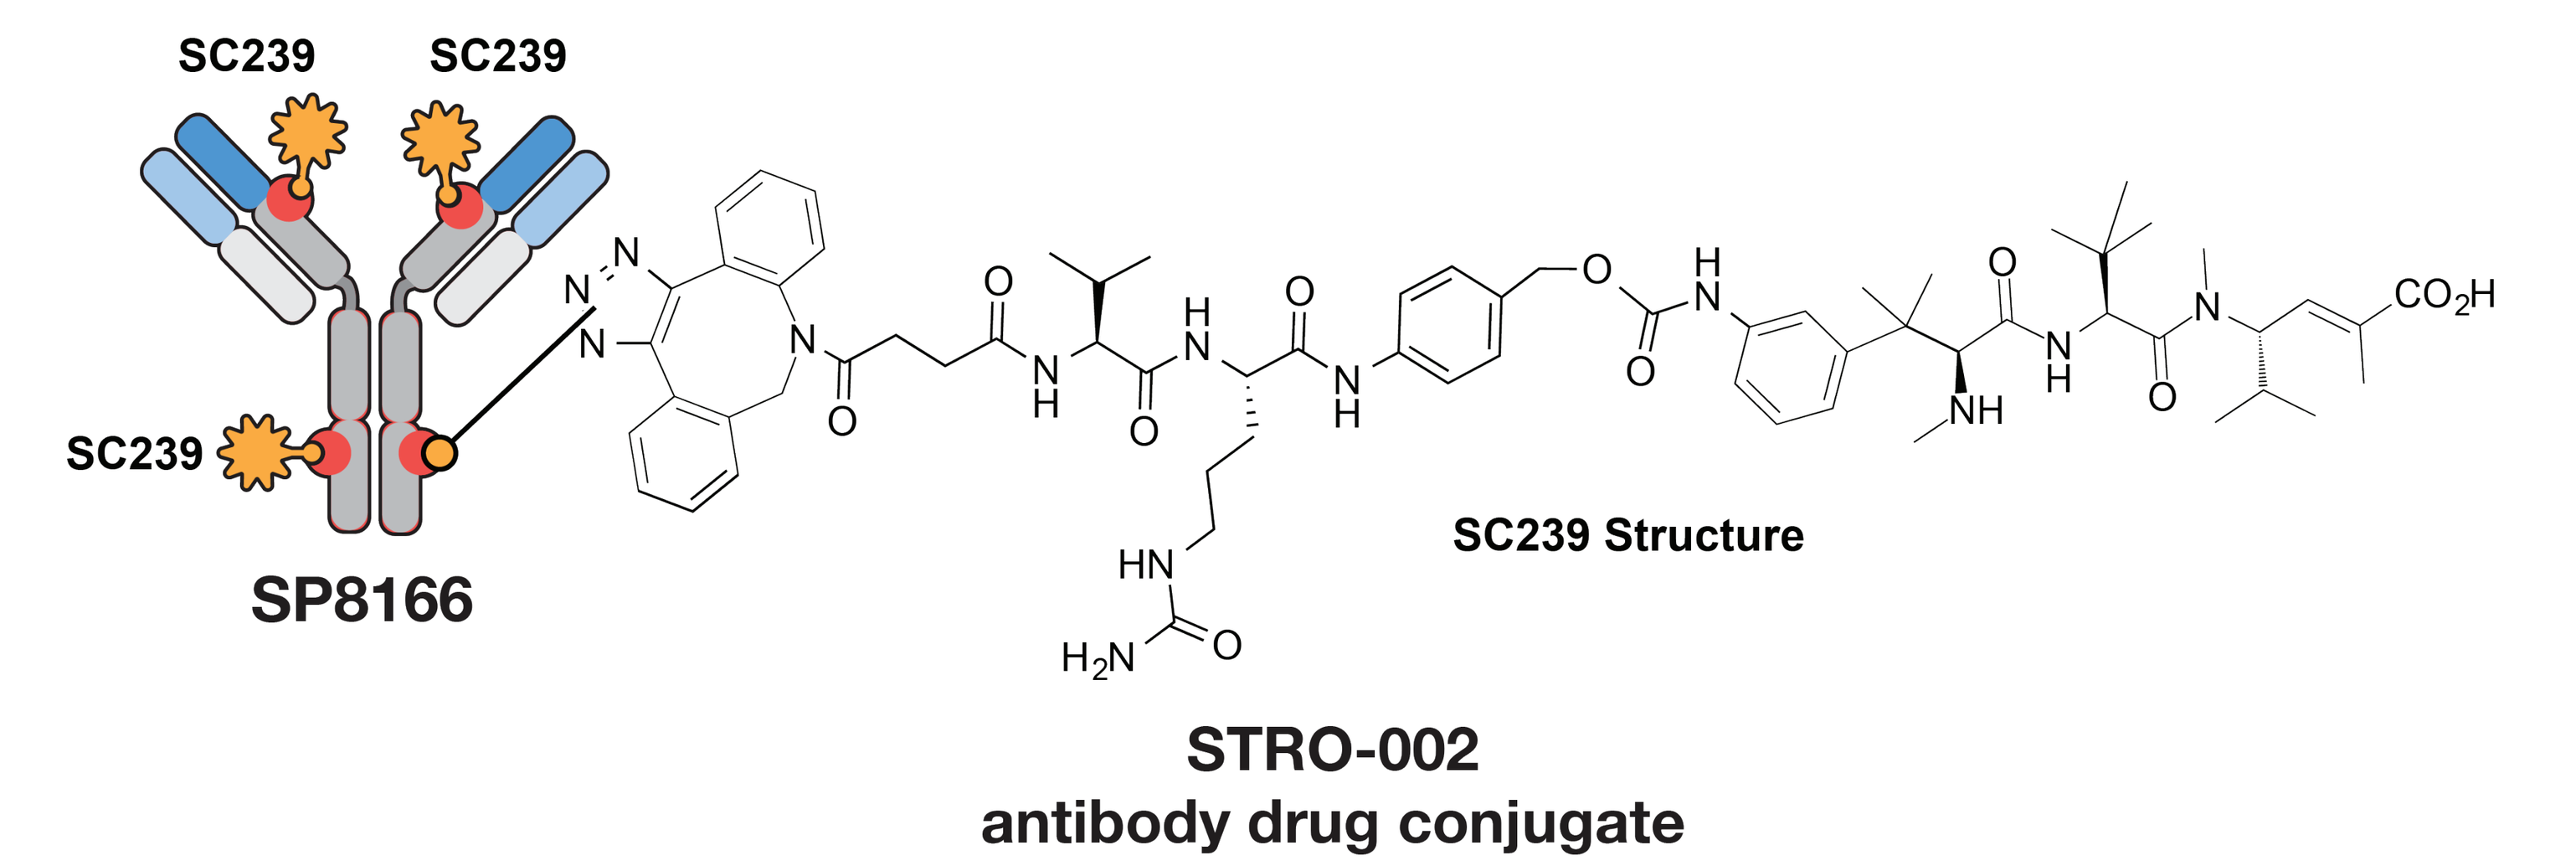
SC239
SC239
SC239
SP8166
SC239 Structure
STRO-002
antibody drug conjugate

Cell Killing Activity of STRO-002 Is Highly Specific for FolRα Expressing Cells

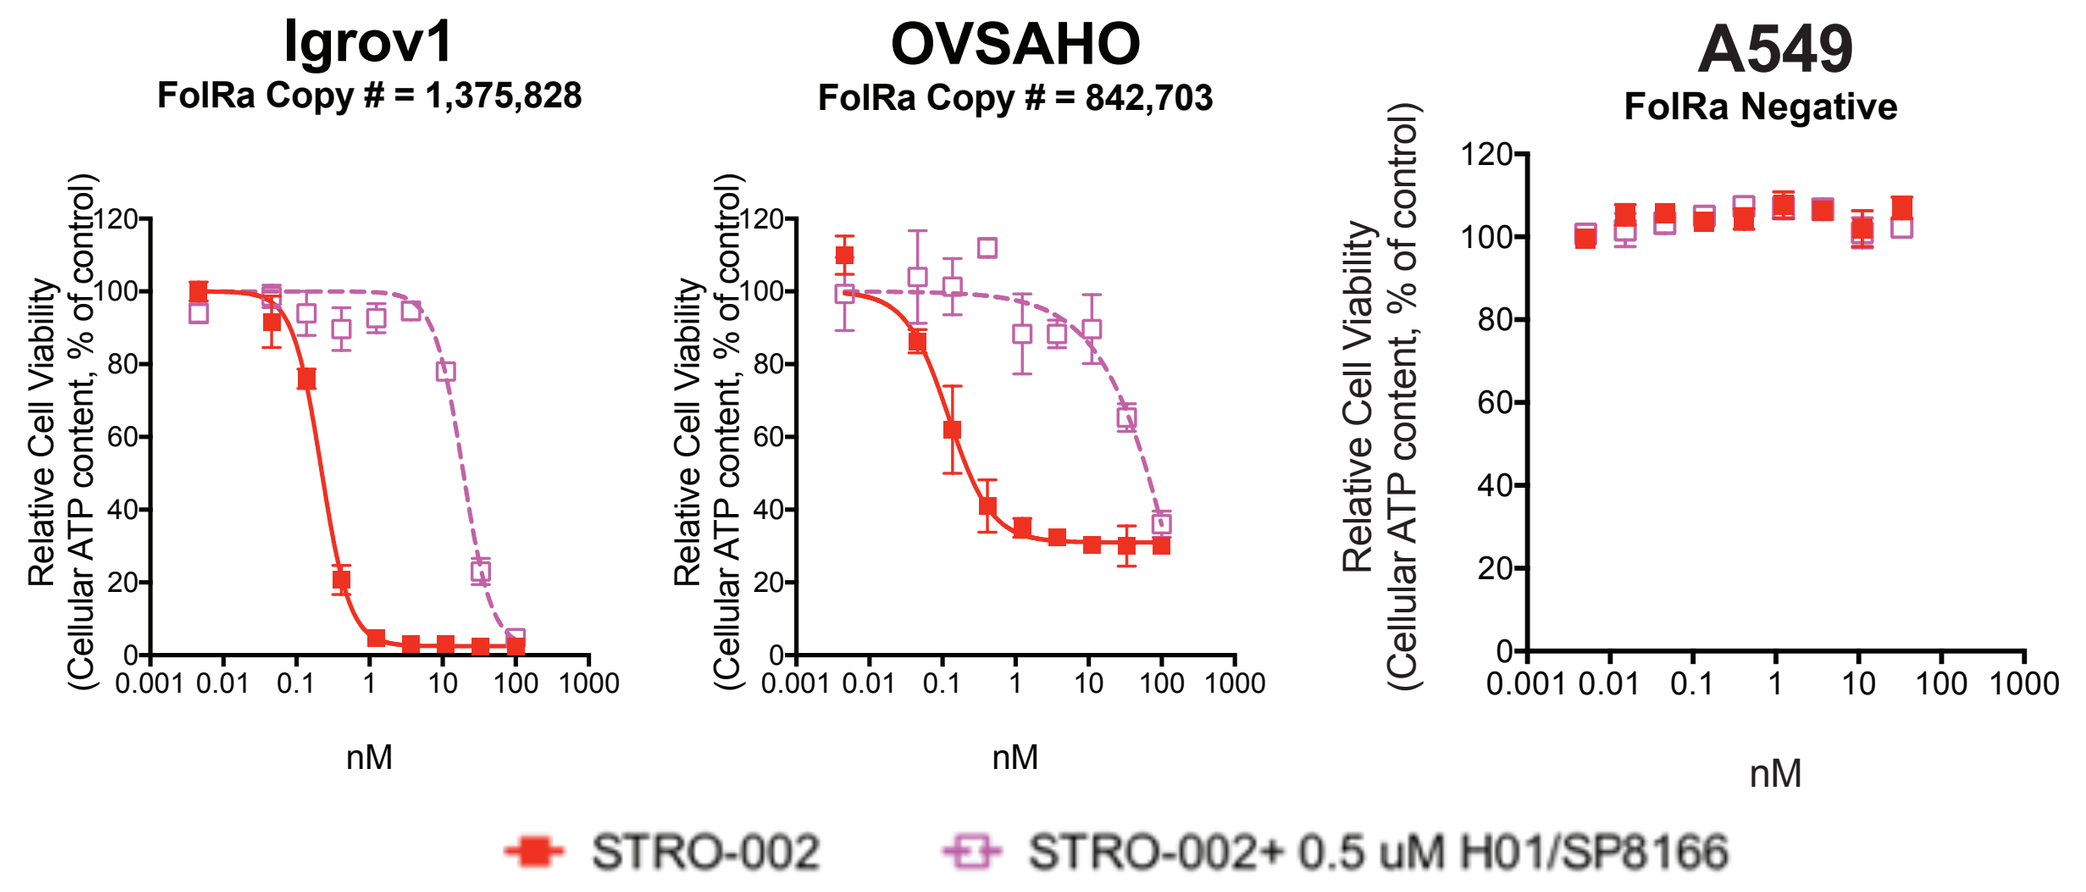
Igrov1
FolRa Copy # = 1,375,828
OVSAHO
FolRa Copy # = 842,703
A549
FolRa Negative
Relative Cell Viability (Cellular ATP content, % of control)
nM
STRO-002
STRO-002+ 0.5 uM H01/SP8166

- STRO-002 showed potent activity in a cell killing assay on FolRα positive Igrov-1 and OVSAHO cells, but no effect on FolRα negative A549 cells.
- Cytotoxic activity of STRO-002 was specific for cells expressing FolRα; and was competed with excess SP8166 (H01).

STRO-002 is Highly Stable *In Vitro* and *In Vivo*

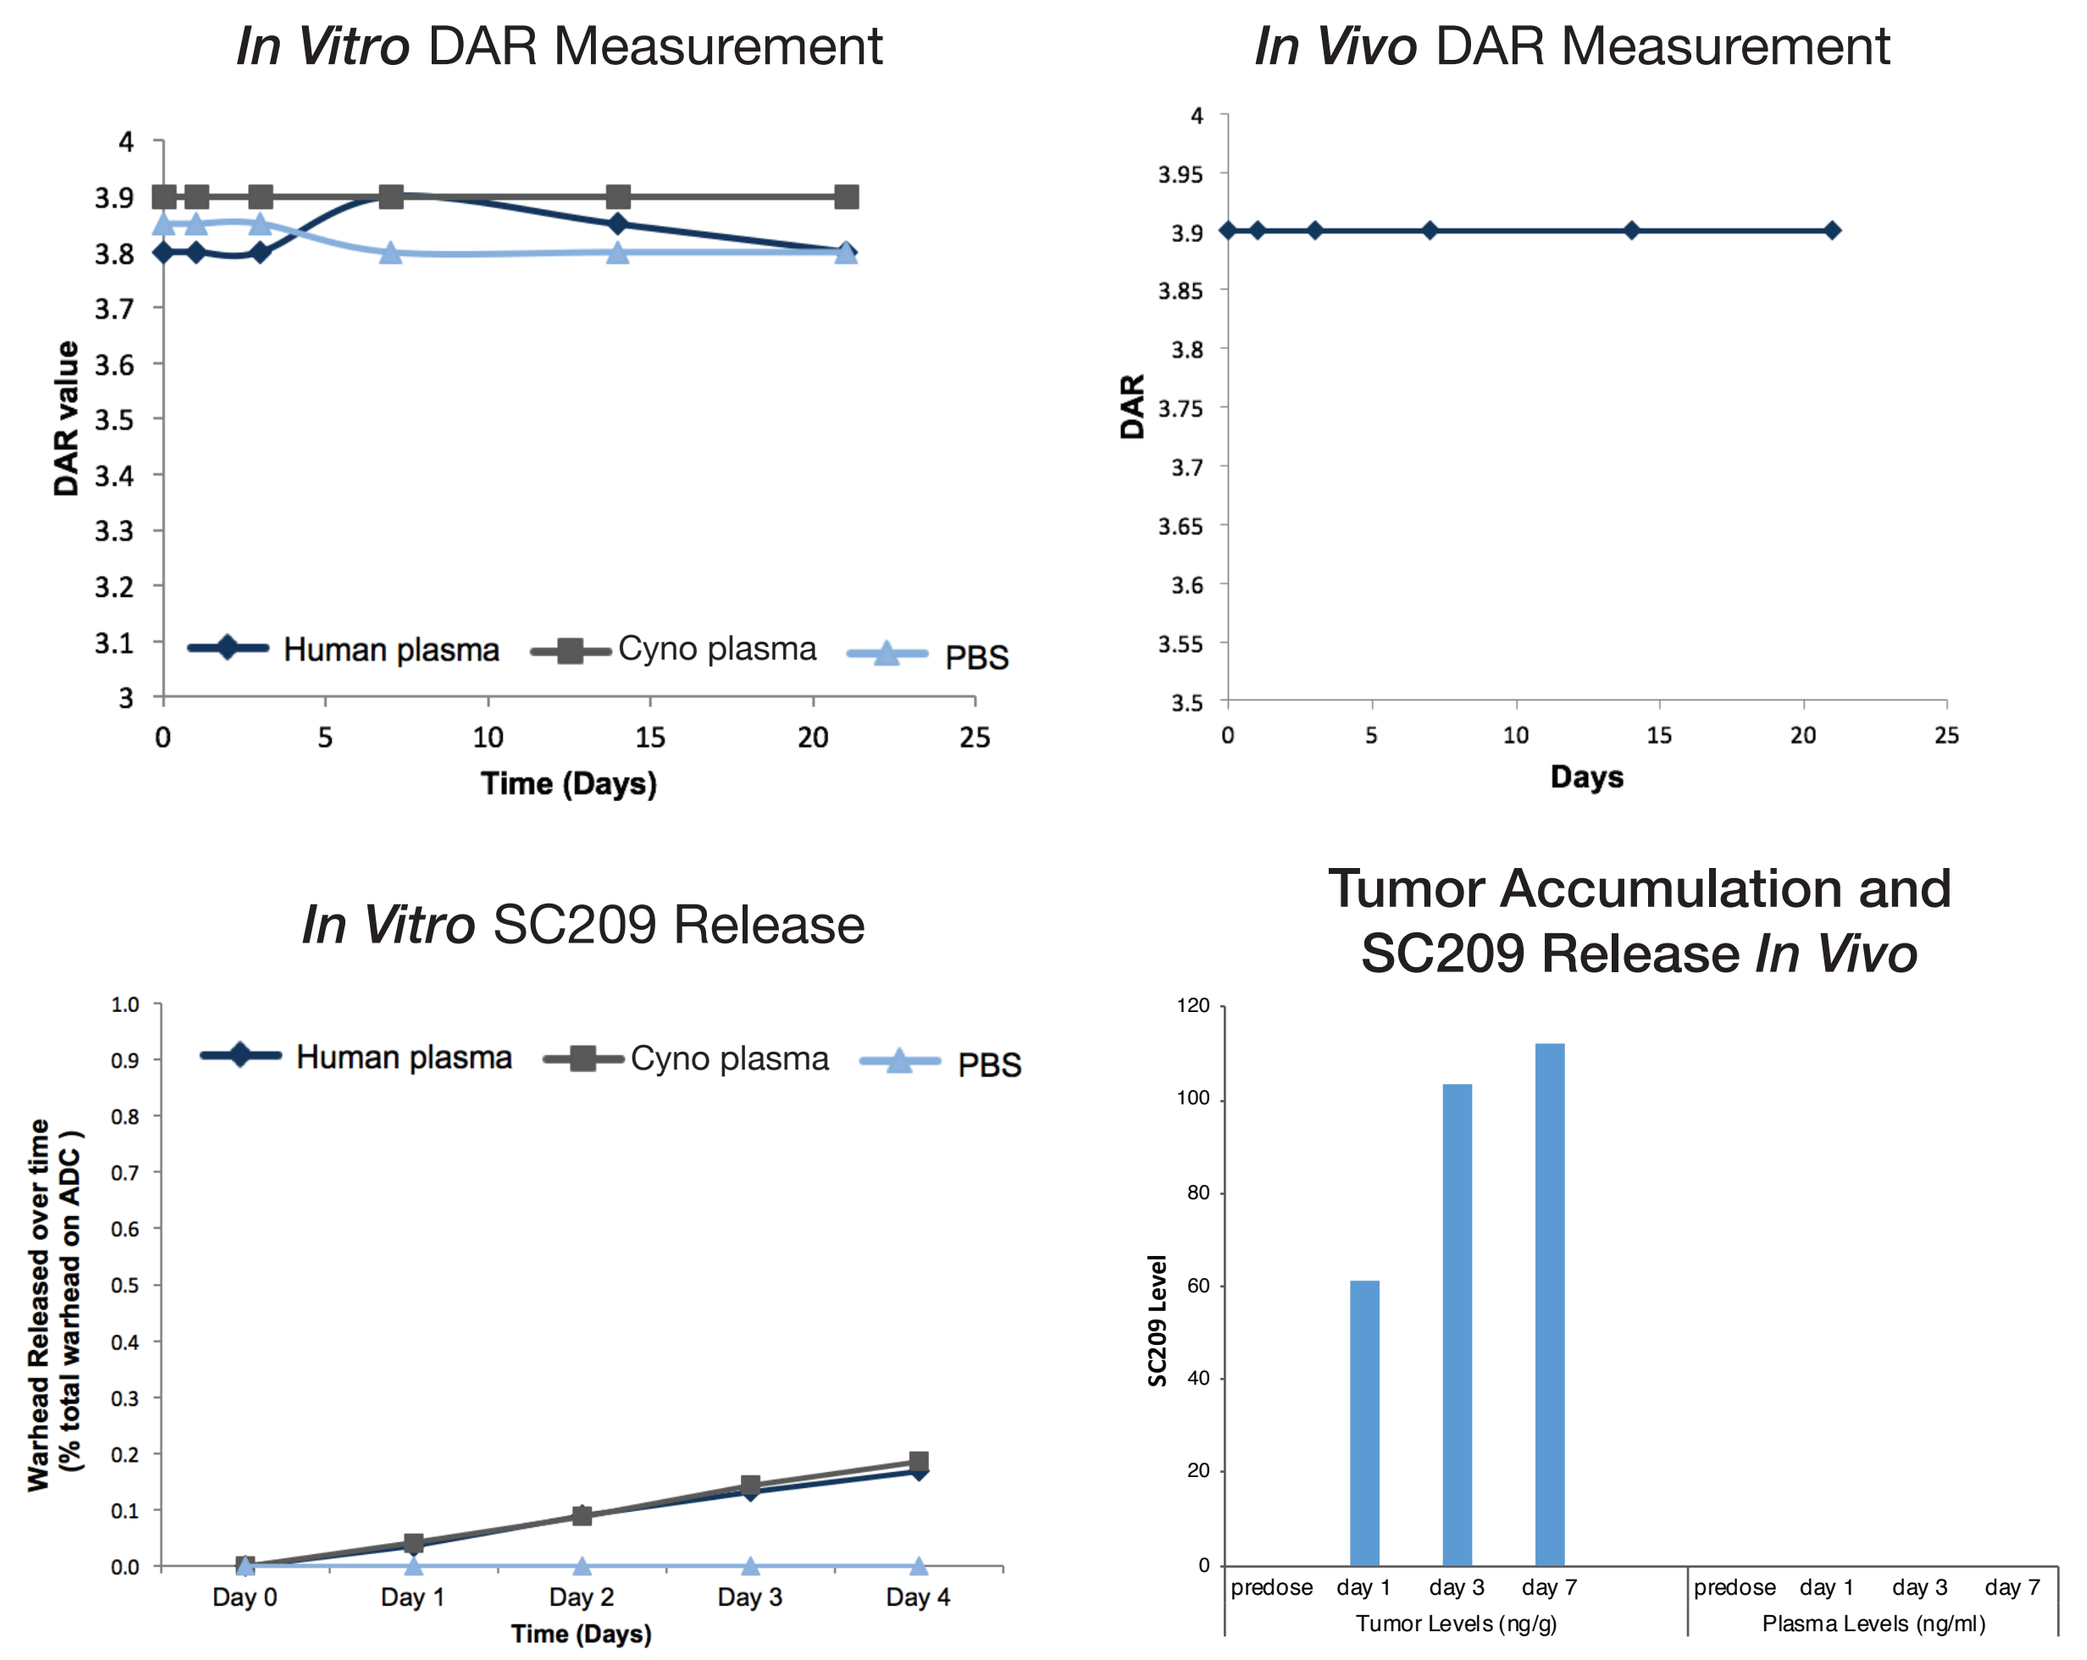
In Vitro DAR Measurement
DAR value
Human plasma
Cyno plasma
PBS
Time (Days)
In Vivo DAR Measurement
DAR
Days
In Vitro SC209 Release
Warhead Released over time (% total warhead on ADC)
Human plasma
Cyno plasma
PBS
Day 0
Day 1
Day 2
Day 3
Day 4
Time (Days)
Tumor Accumulation and SC209 Release In Vivo
SC209 Level
predose
day 1
day 3
day 7
Tumor Levels (ng/g)
Plasma Levels (ng/ml)

- STRO-002 is highly stable *in vitro* (PBS, cynomolgus or human plasma at 50 μg/mL) and *in vivo* (plasma from mice treated with single dose of 5 mg/kg), with a DAR of ~4 being retained until day 21.
- Minimal release of the STRO-002 catabolite, SC209, was observed after a 4-day incubation of STRO-002 in PBS, cyno or human plasma at 100 μg/mL.
- SC209 was accumulated in tumors, but undetectable in circulation of treated mice bearing Igrov1 tumors.
- STRO-002 was isolated from plasma samples by affinity pull-down and DAR measured by LC-MS. Released SC209 was quantitated by LC-MS.

Summary

- We have leveraged Sutro's XpressCF+™ cell-free technology for rapid interrogation and optimization of parameters for a FolRα targeting ADC, including choice of antibody, conjugation sites, DAR, and linker warhead.
- *In vitro* and *in vivo* assessment showed that FolRα ADC composed of antibody H01 conjugated to SC239 at positions Y180 and F404, designated STRO-002, demonstrated the best anti tumor activity in FolRα expressing models, and was selected as the lead antibody for the program.
- STRO-002 demonstrated good pharmacokinetic and pharmacological properties including specificity, stability and safety in cynomolgus monkeys (not shown here), thus making it an ideal candidate for clinical development.
- IND-enabling studies supporting STRO-002 as a potential treatment of FolRα expressing malignancies are on-going and IND submission is planned for the second half of 2018.

References

- Yin, G., et al.; (2012) Aglycosylated antibodies and antibody fragments produced in a scalable in vitro transcription-translation system. mAbs 4:2, 217-225.
- Zimmerman, E. S., et al.; (2014) Production of site-specific antibody–drug conjugates using optimized non-natural amino acids in a cell-free expression system. Bioconjugate Chemistry 2014, 25, 351–361.
- Yin, G., et. al.; (2017) RF1 attenuation enables efficient non-natural amino acid incorporation for production of homogeneous antibody drug conjugates. Scientific Reports 7(1):3026.