749P

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors

Luveltamab tazevibulin, an anti-folate receptor alpha antibody-drug conjugate, in combination with bevacizumab in patients with recurrent high-grade epithelial ovarian cancer: STRO-002-GM2 phase 1 study

Lainie P. Martin[1], Denise S. Uyar[2], Ainhoa Madariaga[3], Alexander I. Spira[4], Ana Oaknin[5], Russell J. Schilder[6], Eva María Guerra[7], Erika P. Hamilton[8], R. Wendel Naumann[9], Domenica Lorusso[10], Lin Lu[11], Craig J. Berman[11], Anne Borgman[11], John W. Moroney[12]

[1]University of Pennsylvania, Abramson Cancer Center, Philadelphia, PA, USA; [2]Froedtert Hospital & Medical College of Wisconsin, Milwaukee, WI, USA; [3]Hospital Universitario 12 de Octubre, Madrid, Spain; [4]Virginia Cancer Specialists, US Oncology Research, Fairfax, VA, USA; [5]Medical Oncology Service, Vall d'Hebron Institute of Oncology, Vall d'Hebron Barcelona Hospital Campus, Barcelona, Spain; [6]Sidney Kimmel Cancer Center, Thomas Jefferson University, Philadelphia, PA, USA; [7]Hospital Ramón y Cajal, Madrid, Spain; [8]Sarah Cannon Research Institute, Nashville, TN, USA; [9]Atrium Health Levine Cancer Institute, Charlotte, NC, USA; [10]Fondazione Policlinico Universitario A Gemelli IRCCS, Rome, Italy; [11]Sutro Biopharma, Inc., South San Francisco, CA, USA; [12]University of Chicago, Chicago, IL, USA

INTRODUCTION

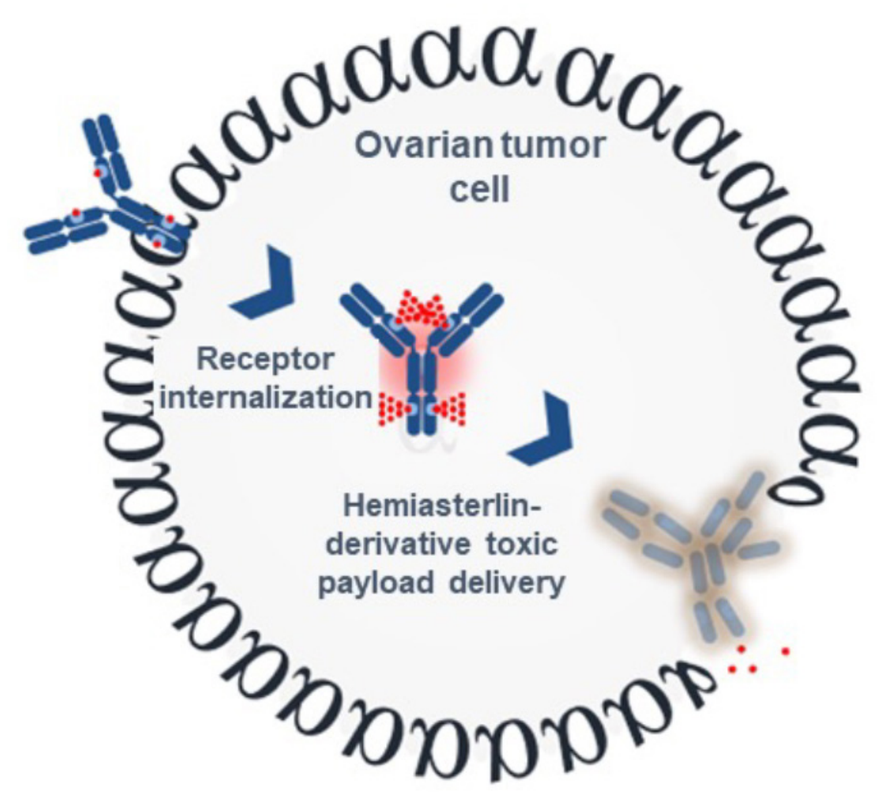
Ovarian tumor cell
Receptor internalization
Hemiasterlin-derivative toxic payload delivery

- Folate receptor alpha (FRα) is overexpressed in epithelial ovarian cancer (EOC), making it a favorable therapeutic target for recurrent EOC, an area of high unmet need, with limited disease control and survival in the platinum-resistant setting[1,2]
- **Luveltamab tazevibulin (luvelta; STRO-002)** is a FRα antibody-drug conjugate with a stable cleavable linker and a 3-aminophenyl hemiasterlin warhead that induces cytotoxic and immunogenic cell death[3]
- Luvelta monotherapy demonstrated encouraging antitumor activity and a manageable safety profile in the phase 1, open-label STRO-002-GM1 (NCT03748186) study in patients with recurrent EOC[4]
- Luvelta combined with bevacizumab (bev) has shown increased antitumor effects (vs luvelta alone) in ovarian cancer xenograft models, and preliminary clinical data suggest increased activity of the combination[5]
- Herein, we report the outcomes from the escalation portion of the ongoing phase 1/1b trial (NCT05200364) evaluating the recommended phase 2 dose (RP2D), safety, and antitumor activity of luvelta in combination with bev in patients with recurrent EOC
- Expansion at RP2D is ongoing; an additional 23 patients have been enrolled to date

METHODS

- STRO-002-GM2 is a global phase 1/2a, open-label, multicenter, dose-escalation and dose-expansion study including sites in US, Spain, Italy, and France (planned)
- Dose escalation employed a standard 3+3 design, with a 21-day dose-limiting toxicity (DLT) assessment period to determine the RP2D (**Figure 1**) for evaluation in dose expansion
- FRα expression data were not required for study entry; expression was determined retrospectively by immunohistochemistry, using VENTANA FOLR1 [FOLR1-2.1] RxDx Assay (Ventana Medical Systems, Roche Labs)
- Primary endpoints are incidence and severity of treatment-emergent adverse events (TEAEs), DLTs, RP2D, and overall response rate (ORR)

Figure 1. Study Schema

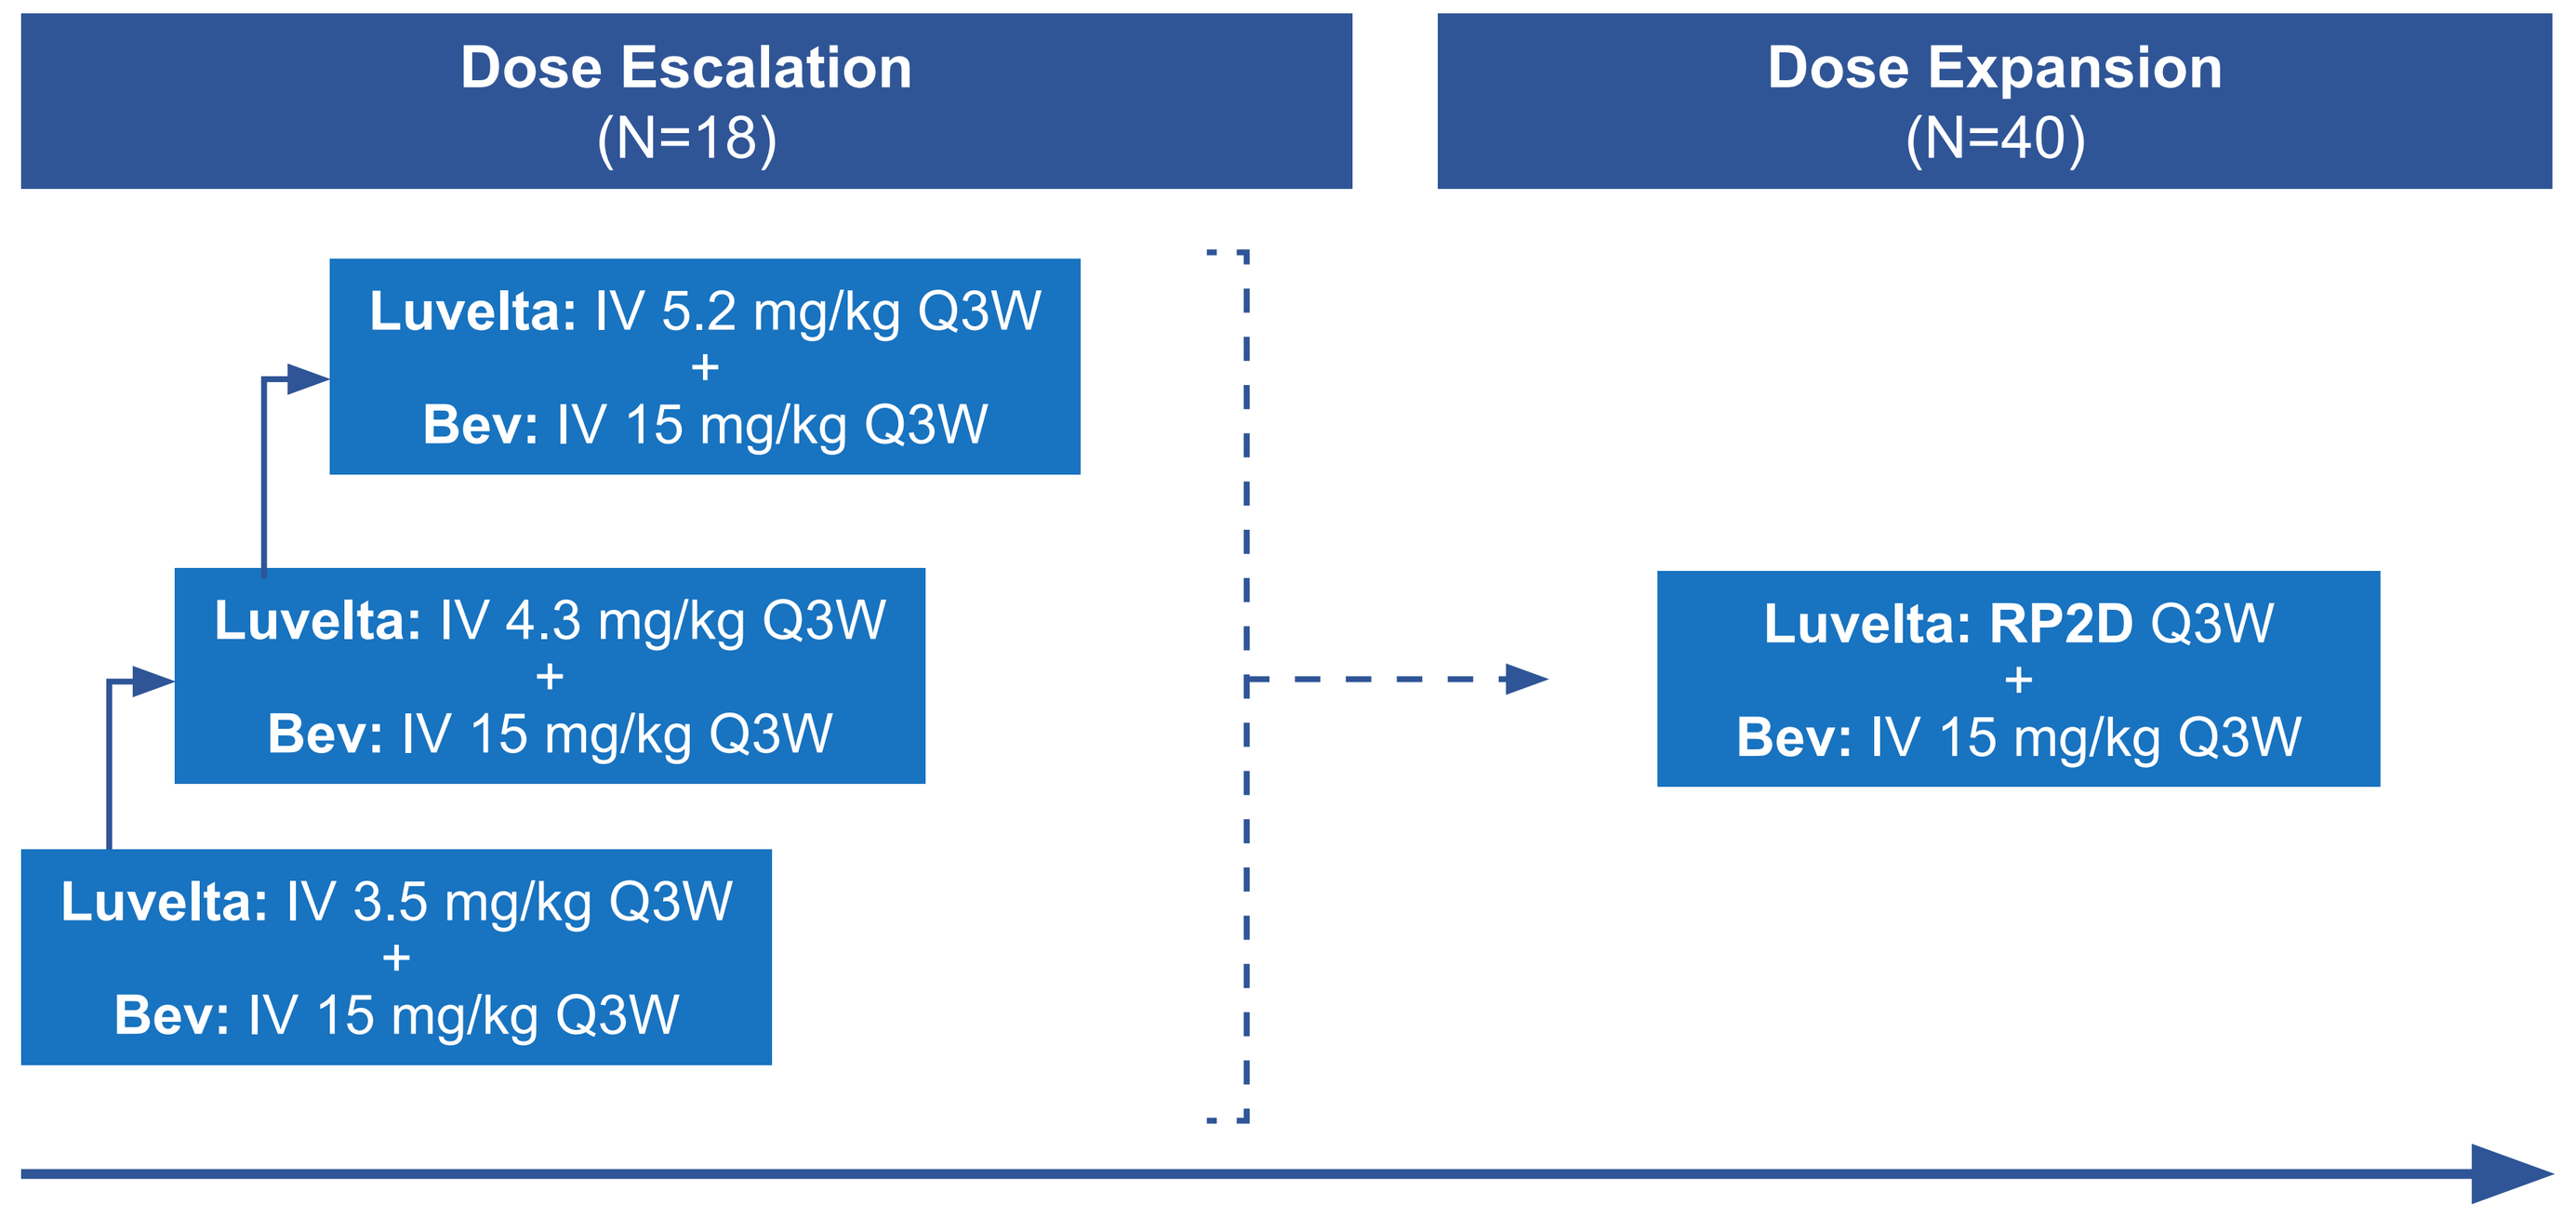
Dose Escalation
(N=18)
Dose Expansion
(N=40)
Luvelta: IV 5.2 mg/kg Q3W + Bev: IV 15 mg/kg Q3W
Luvelta: IV 4.3 mg/kg Q3W + Bev: IV 15 mg/kg Q3W
Luvelta: IV 3.5 mg/kg Q3W + Bev: IV 15 mg/kg Q3W
Luvelta: RP2D Q3W + Bev: IV 15 mg/kg Q3W

Bev, bevacizumab; IV, intravenous; Luvelta, luveltamab tazevibulin; Q3W, every 3 weeks; RP2D, recommended phase 2 dose.

Key Inclusion Criteria

- Adults (≥18 years) with ECOG PS 0-1, high-grade serous EOC, fallopian tube, or primary peritoneal cancer, and RECIST v1.1 evaluable disease
- Relapse/progression on the last treatment and 1 of the following:
 - a) primary platinum refractory and received ≤2 prior regimens;
 - b) platinum resistant and received 1-4 prior regimens; or
 - c) platinum sensitive and received 2-4 prior regimens

ECOG, Eastern Cooperative Oncology Group; EOC, epithelial ovarian cancer; PS, performance status; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1.

PATIENTS

Figure 2. Patient Distribution

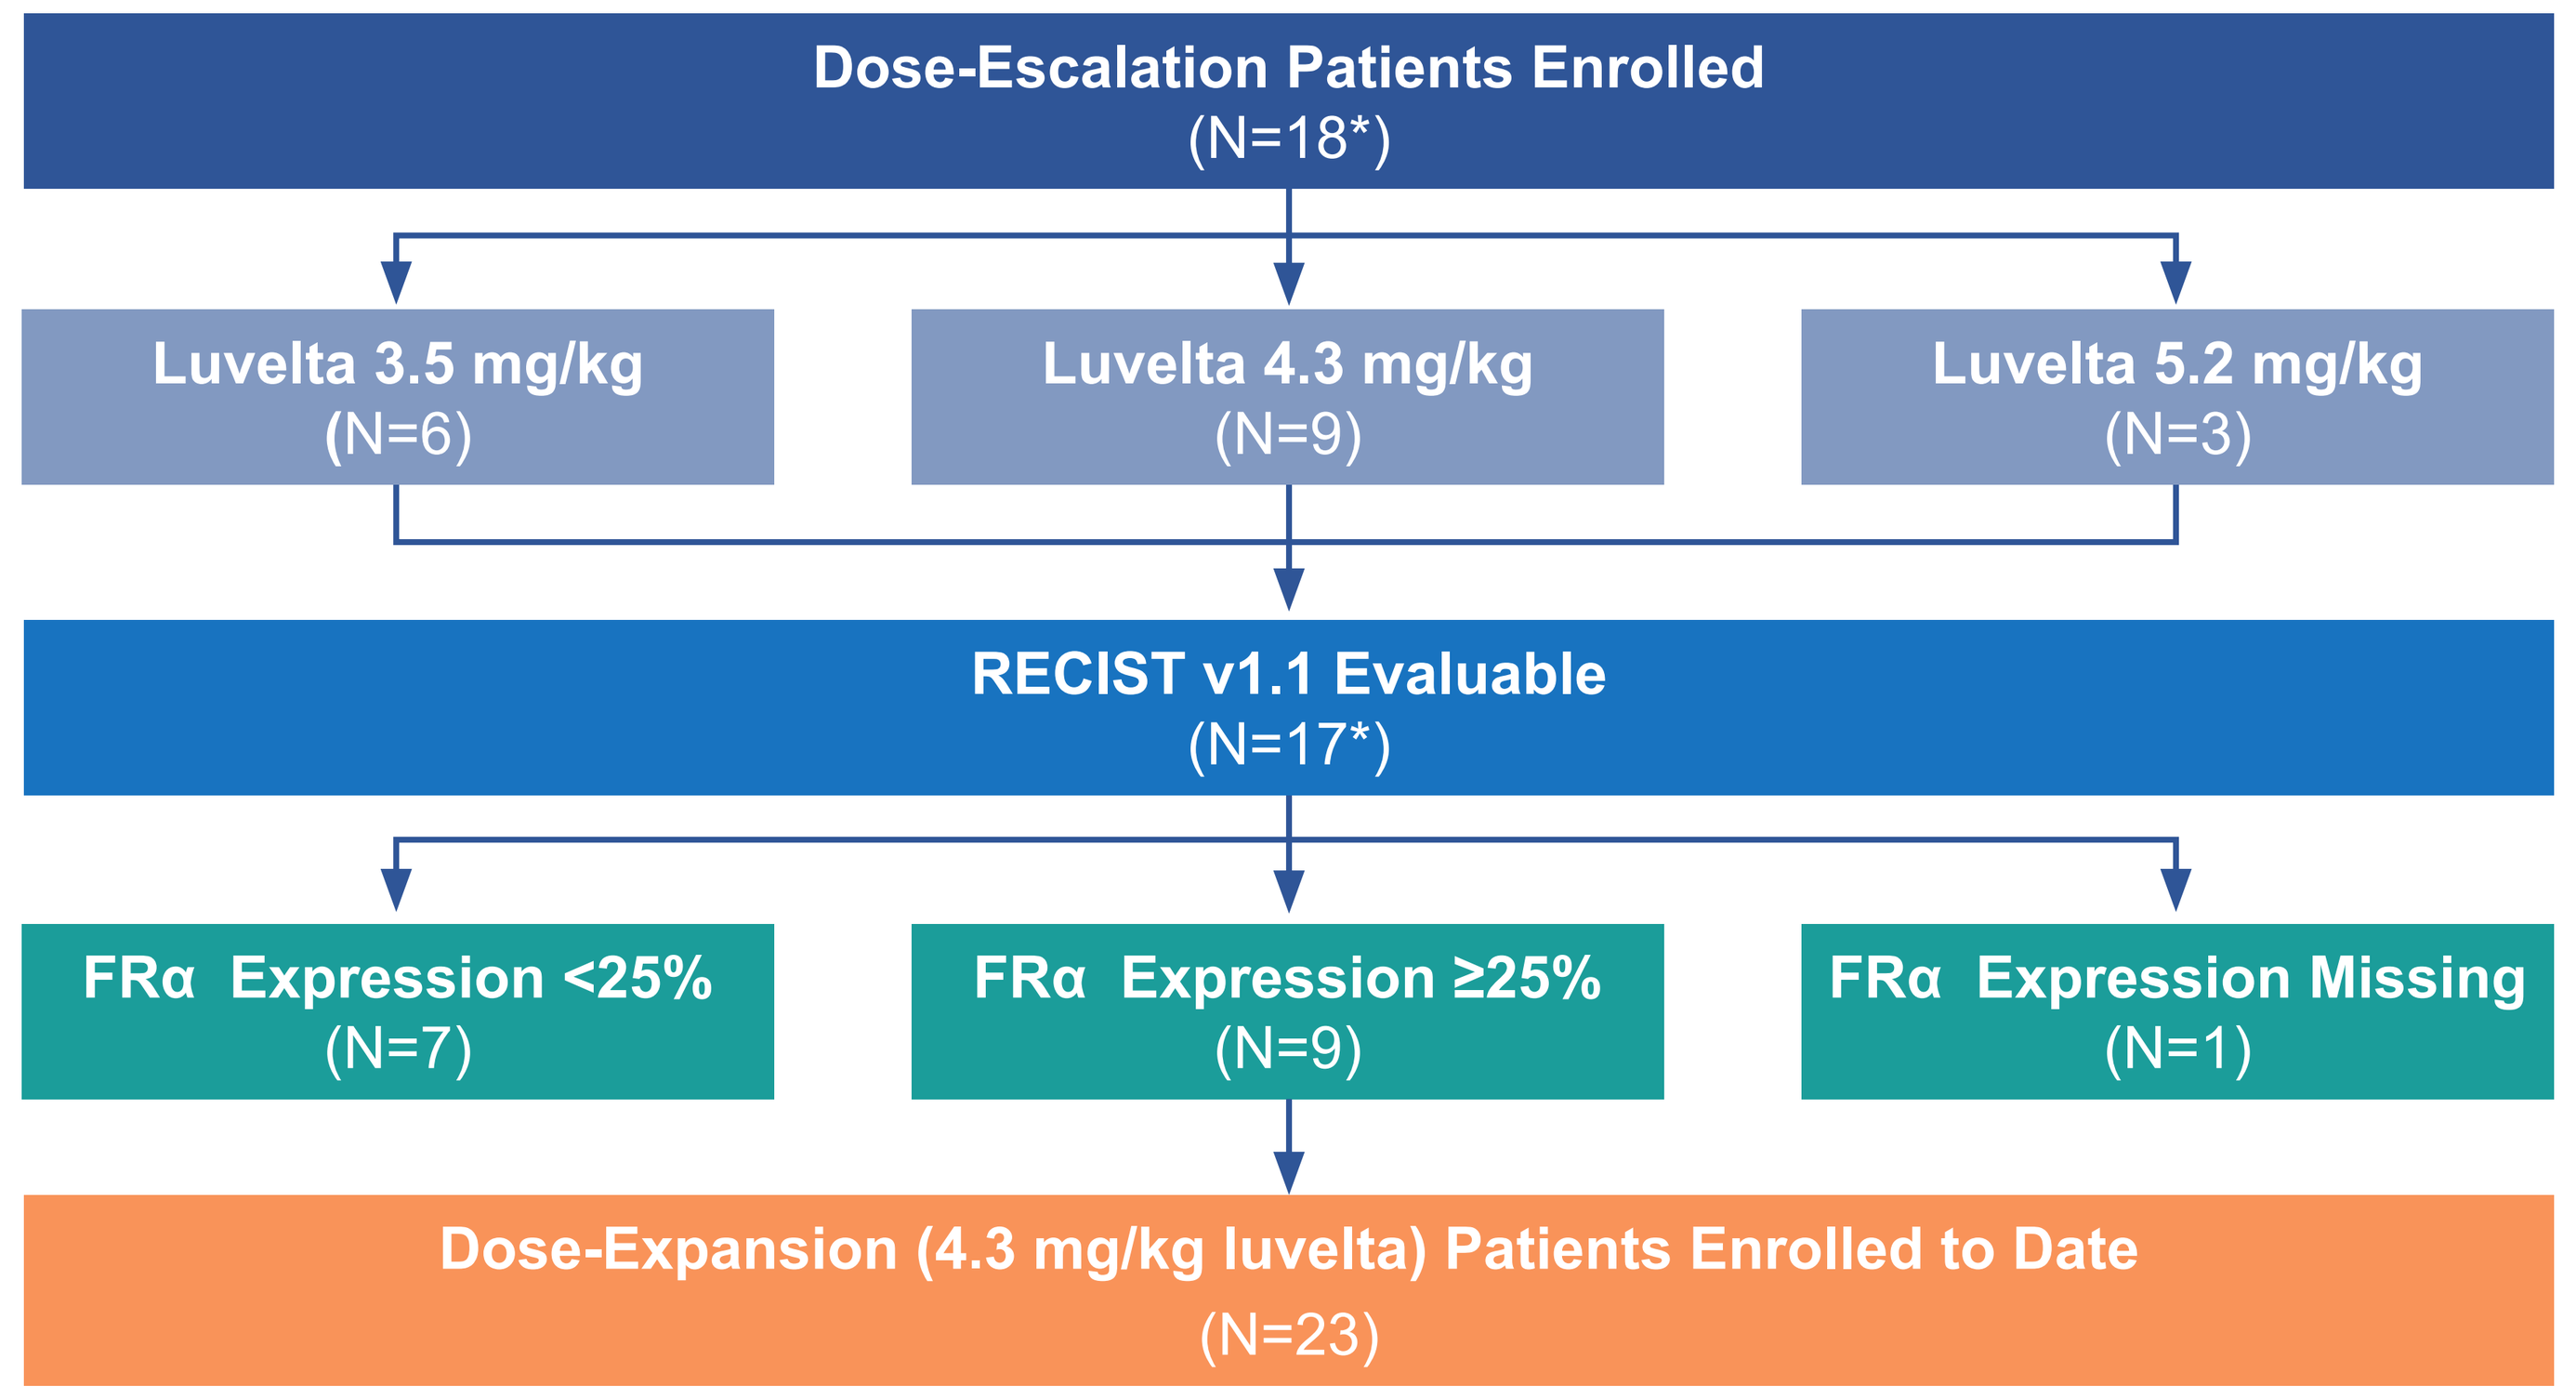
Dose-Escalation Patients Enrolled
(N=18*)
Luvelta 3.5 mg/kg
(N=6)
Luvelta 4.3 mg/kg
(N=9)
Luvelta 5.2 mg/kg
(N=3)
RECIST v1.1 Evaluable
(N=17*)
FRα Expression <25%
(N=7)
FRα Expression ≥25%
(N=9)
FRα Expression Missing
(N=1)
Dose-Expansion (4.3 mg/kg luvelta) Patients Enrolled to Date
(N=23)

Data cut off date: August 2nd, 2024.
*One patient from the dose-escalation phase remains on treatment.
FRα, folate receptor alpha; luvelta, luveltamab tazevibulin; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1.

Patient Demographics and Disease Characteristics (N=18)	
Median age, years (range)	63 (47–74)
ECOG PS, n (%)	
0	10 (56)
1	8 (44)
Median number of prior therapies (range)	2 (1–4)
Prior bev, n (%)	10 (56)
Prior PARPi, n (%)	13 (72)
Prior platinum, n (%)	18 (100)
BRCA positive, n (%)	2 (11)
Ascites, n (%)	5 (28)
PSOC	3 (17)

Data cut off date: August 2nd, 2024.
Bev, bevacizumab; *BRCA*, BReast CAncer gene; ECOG, Eastern Cooperative Oncology Group; PARPi, poly (ADP-ribose) polymerase inhibitor; PS, performance status; PSOC, platinum-sensitive ovarian cancer.

SAFETY

- The 4.3 mg/kg dose was selected for evaluation in the expansion phase
- At the 5.2 mg/kg dose, 1 out of 3 patients enrolled experienced a DLT of grade 3 nausea and a DLT of grade 4 decreased appetite on C1D11
- 9 (50%) patients experienced a TEAE leading to luvelta dose reduction; the most frequent TEAE leading to dose reduction was neutropenia
- 8 (44%) patients experienced treatment-related AEs leading to luvelta discontinuation
- Two deaths occurred in the dose-escalation phase:
 - Grade 5 non-neutropenic sepsis (C2): assessed as doubtfully related to luvelta and possibly related to bev; the probable cause of sepsis was malignant bowel perforation caused by progressive disease
 - Grade 5 sepsis (C12): occurred after the patient underwent a diabetic foot ulcer drainage procedure; considered not drug-related by the investigator (related to skin infection), but the sponsor upgraded attribution to possibly related
 - Safety guidelines were updated to advise that patients be evaluated by their oncologist prior to undergoing any surgical procedure

SAFETY

n, (%)	Any Grade	Grade ≥3
TEAEs >25% Incidence (Any Grade and Grade ≥3) (N=18)		
Patients with ≥1 TEAE	18 (100)	13 (72)
Neutropenia[a]	13 (72)	8[b] (44)
Constipation	11 (61)	1 (6)
Nausea	11 (61)	1 (6)
Arthralgia	11 (61)	0
Asthenia	8 (44)	2 (11)
Abdominal pain	7 (39)	1 (6)
Diarrhea	7 (39)	0
Fatigue	7 (39)	0
Aspartate aminotransferase increased	6 (33)	0
Headache	6 (33)	0
Thrombocytopenia	5 (28)	1 (6)
Vomiting	5 (28)	1 (6)
Platelet count decreased	5 (28)	1 (6)
Myalgia	5 (28)	0

Data cut off date: August 2nd, 2024.
[a]Neutropenia included the following preferred terms: neutropenia, febrile neutropenia, and neutrophil count decrease. [b]Of the 8 grade ≥3 neutropenia events, 1 event was febrile neutropenia.
TEAE, treatment-emergent adverse event.

EFFICACY

- **Figure 3** summarizes the response outcomes in the dose escalation, showing antitumor activity in all groups of patients
- An ORR of 35.3% (95% CI: 14.2, 61.7) was observed in the overall population with a median duration of response of 9.3 months (95%CI: 5.1, not reached)
- At the RP2D (4.3 mg/kg), 5 of 9 patients had a response; no patients had a response at 3.5 mg/kg and 1 (50%) patient had a response at 5.2 mg/kg

Figure 3. Response Outcomes in Dose Escalation

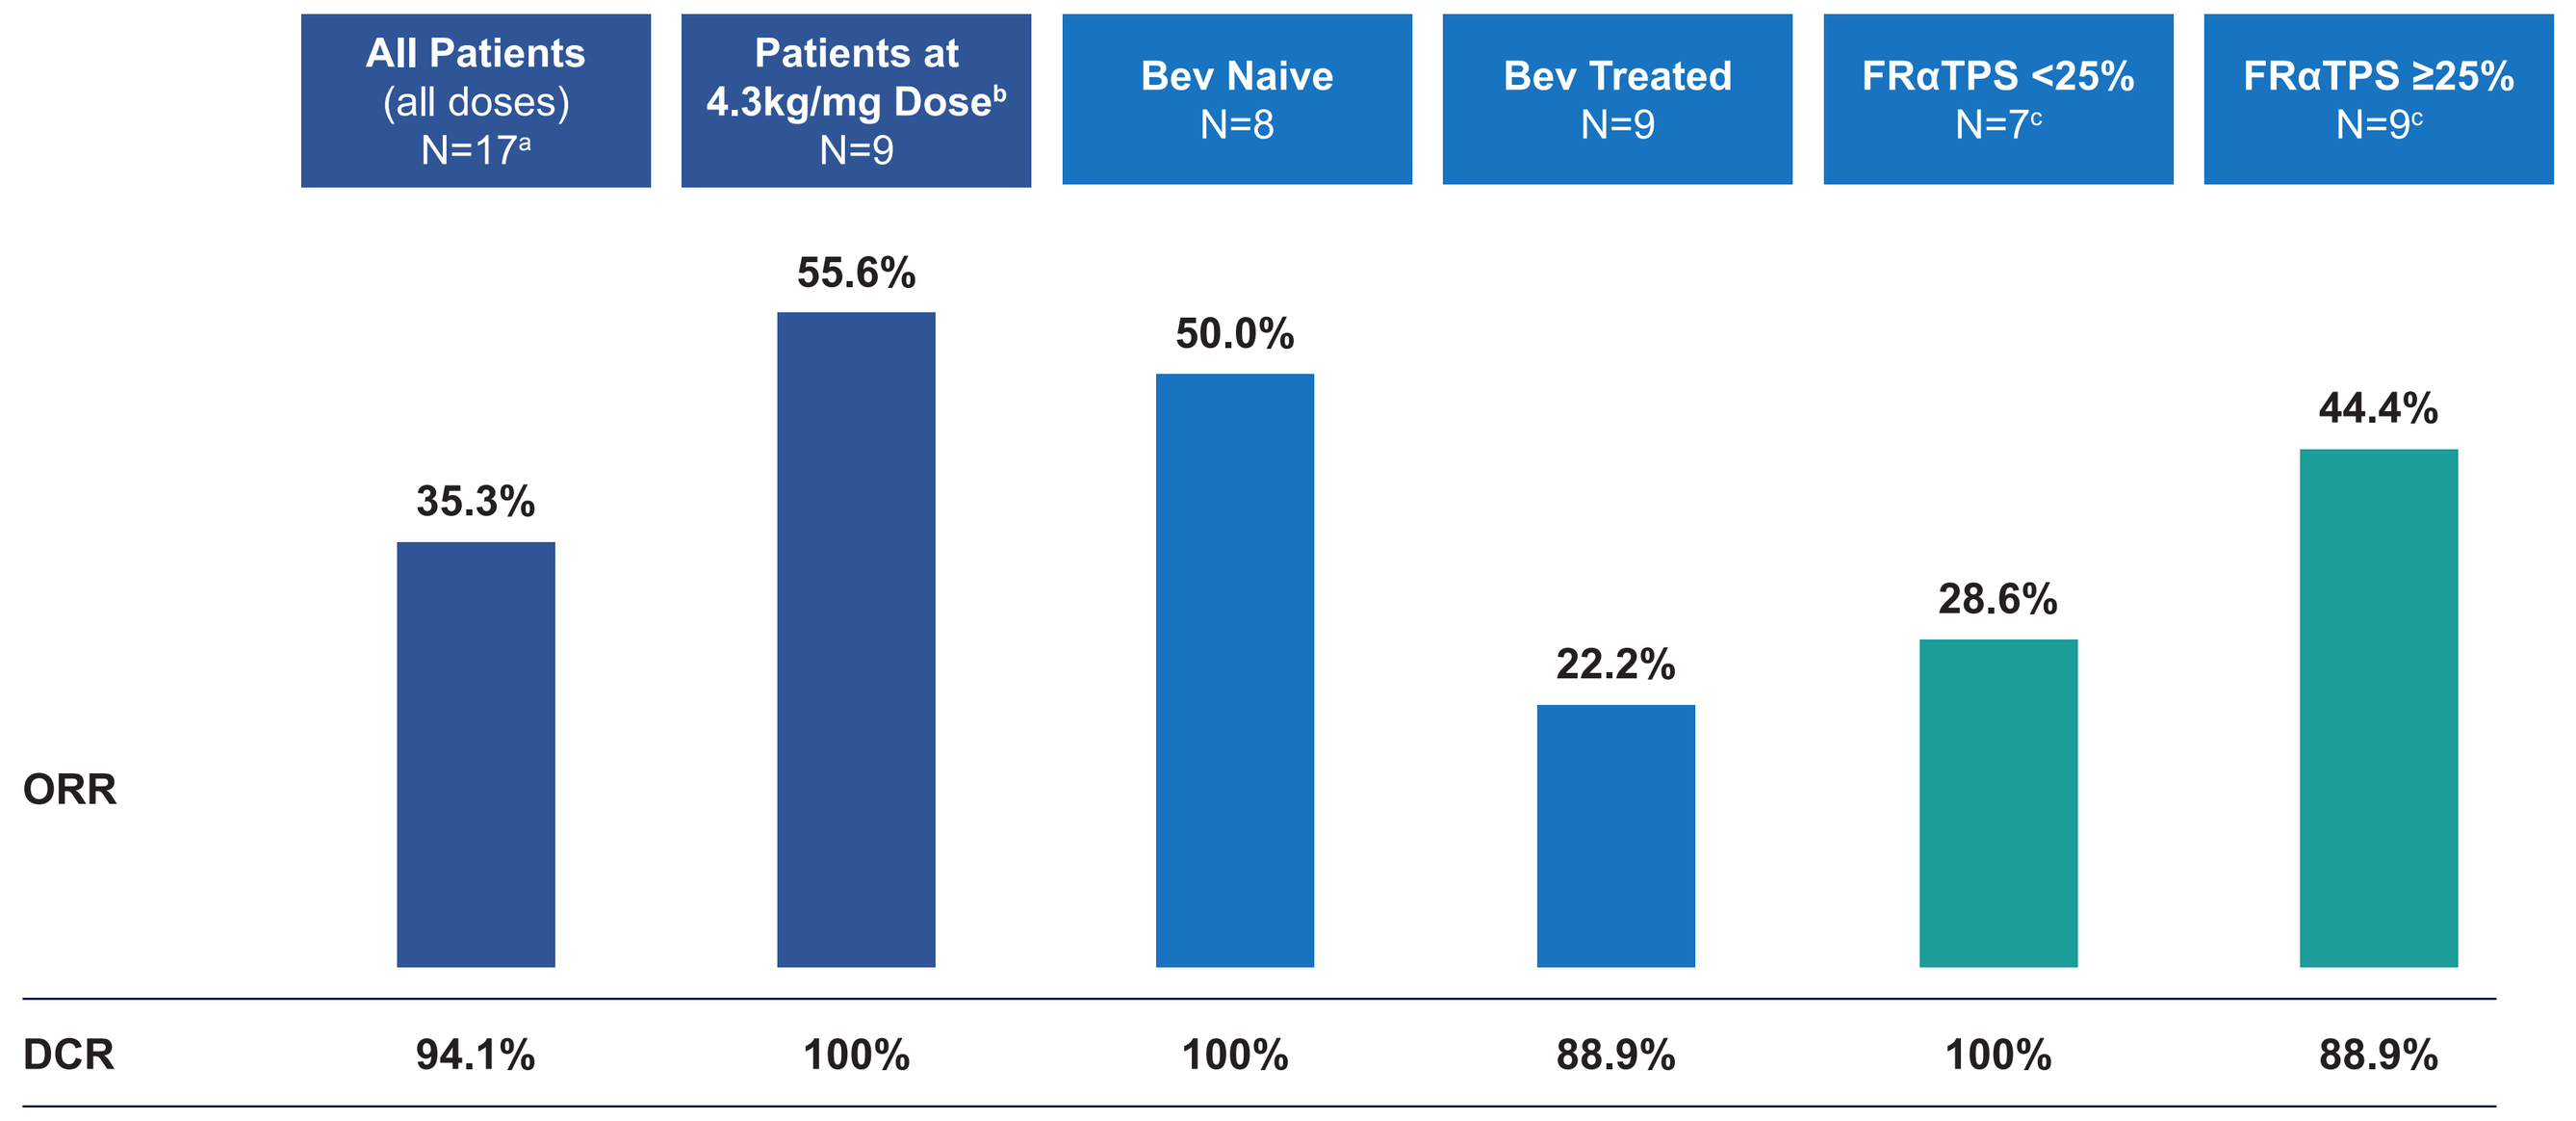
All Patients (all doses) N=17[a]
Patients at 4.3kg/mg Dose[b] N=9
Bev Naive N=8
Bev Treated N=9
FRαTPS <25% N=7[c]
FRαTPS ≥25% N=9[c]
ORR
35.3%
55.6%
50.0%
22.2%
28.6%
44.4%
DCR
94.1%
100%
100%
88.9%
100%
88.9%

Data cut off date: August 2nd, 2024.
[a]One patient was not evaluable for response; [b]Selected RP2D; [c]FRα expression missing for 1 patient.
Responses were assessed in patients with baseline and post-treatment assessments (N=17).
Bev, bevacizumab; CI, confidence interval; DCR, disease control rate; FRα, folate receptor alpha; ORR, overall response rate; RP2D, recommended phase 2 dose; TPS, tumor proportion score.

EFFICACY

Figure 4. Duration of Treatment and Responses (N=18)

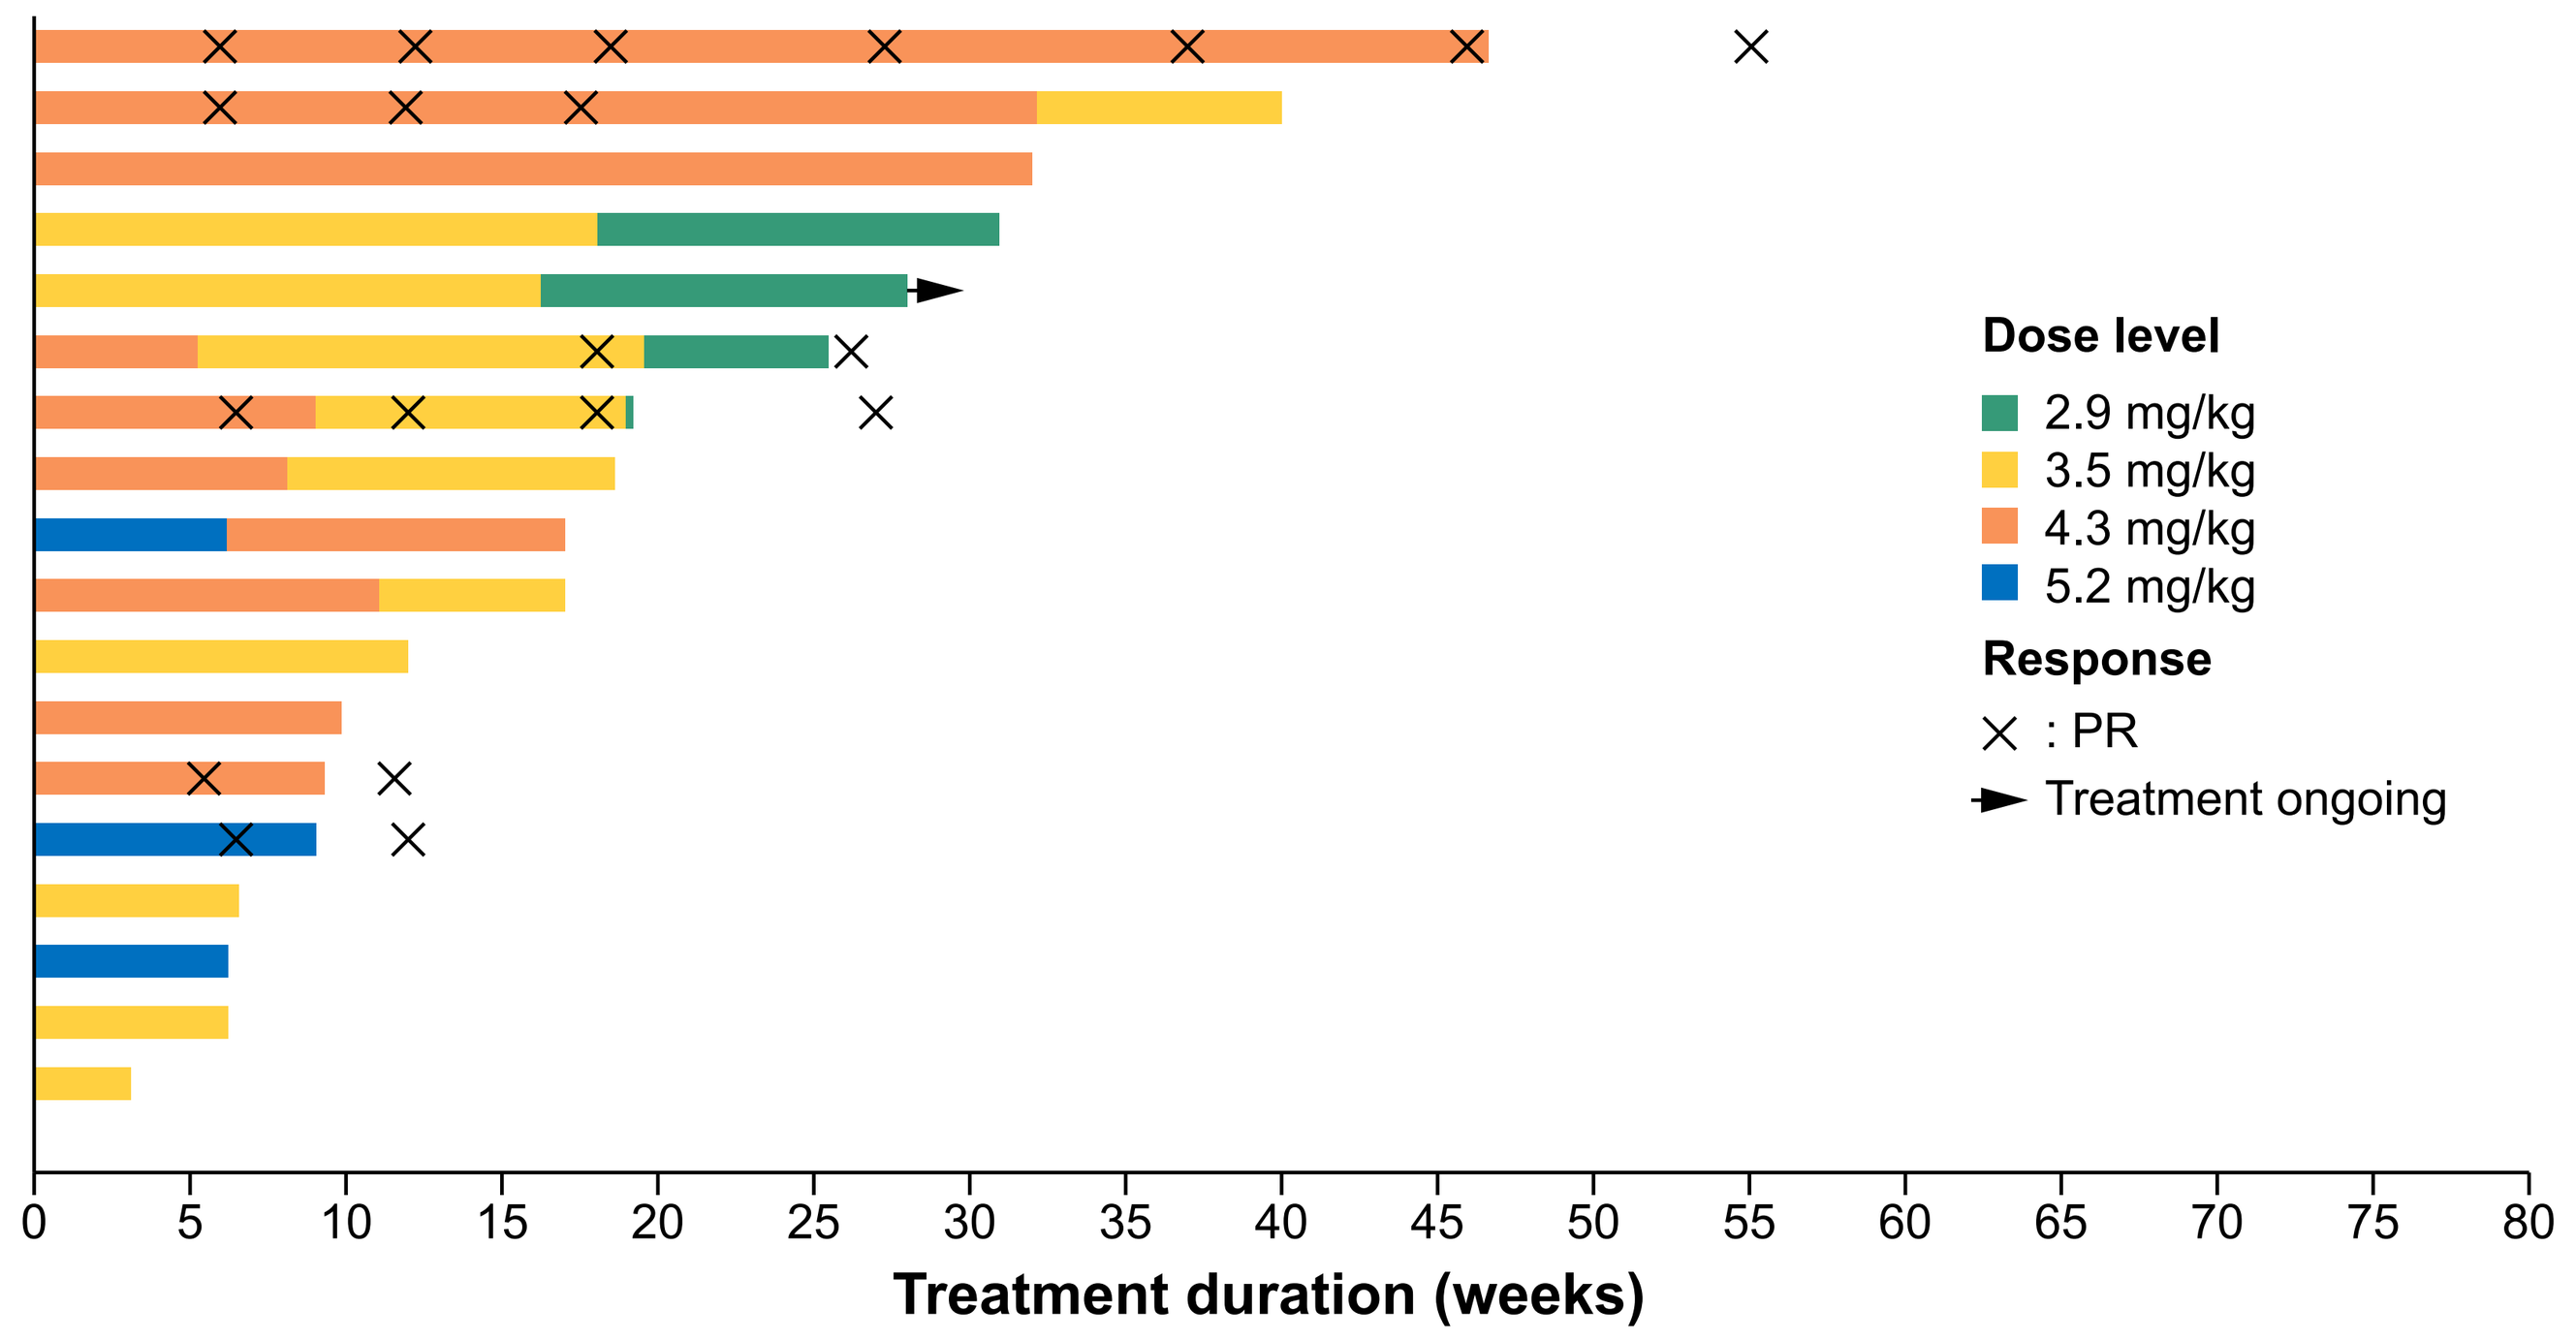
Dose level
2.9 mg/kg
3.5 mg/kg
4.3 mg/kg
5.2 mg/kg
Response
: PR
Treatment ongoing
0
5
10
15
20
25
30
35
40
45
50
55
60
65
70
75
80
Treatment duration (weeks)

Data cut off date: August 2nd, 2024.
PR, partial response.

Figure 5. Maximum Reduction in Target Tumor Lesions (N=17)

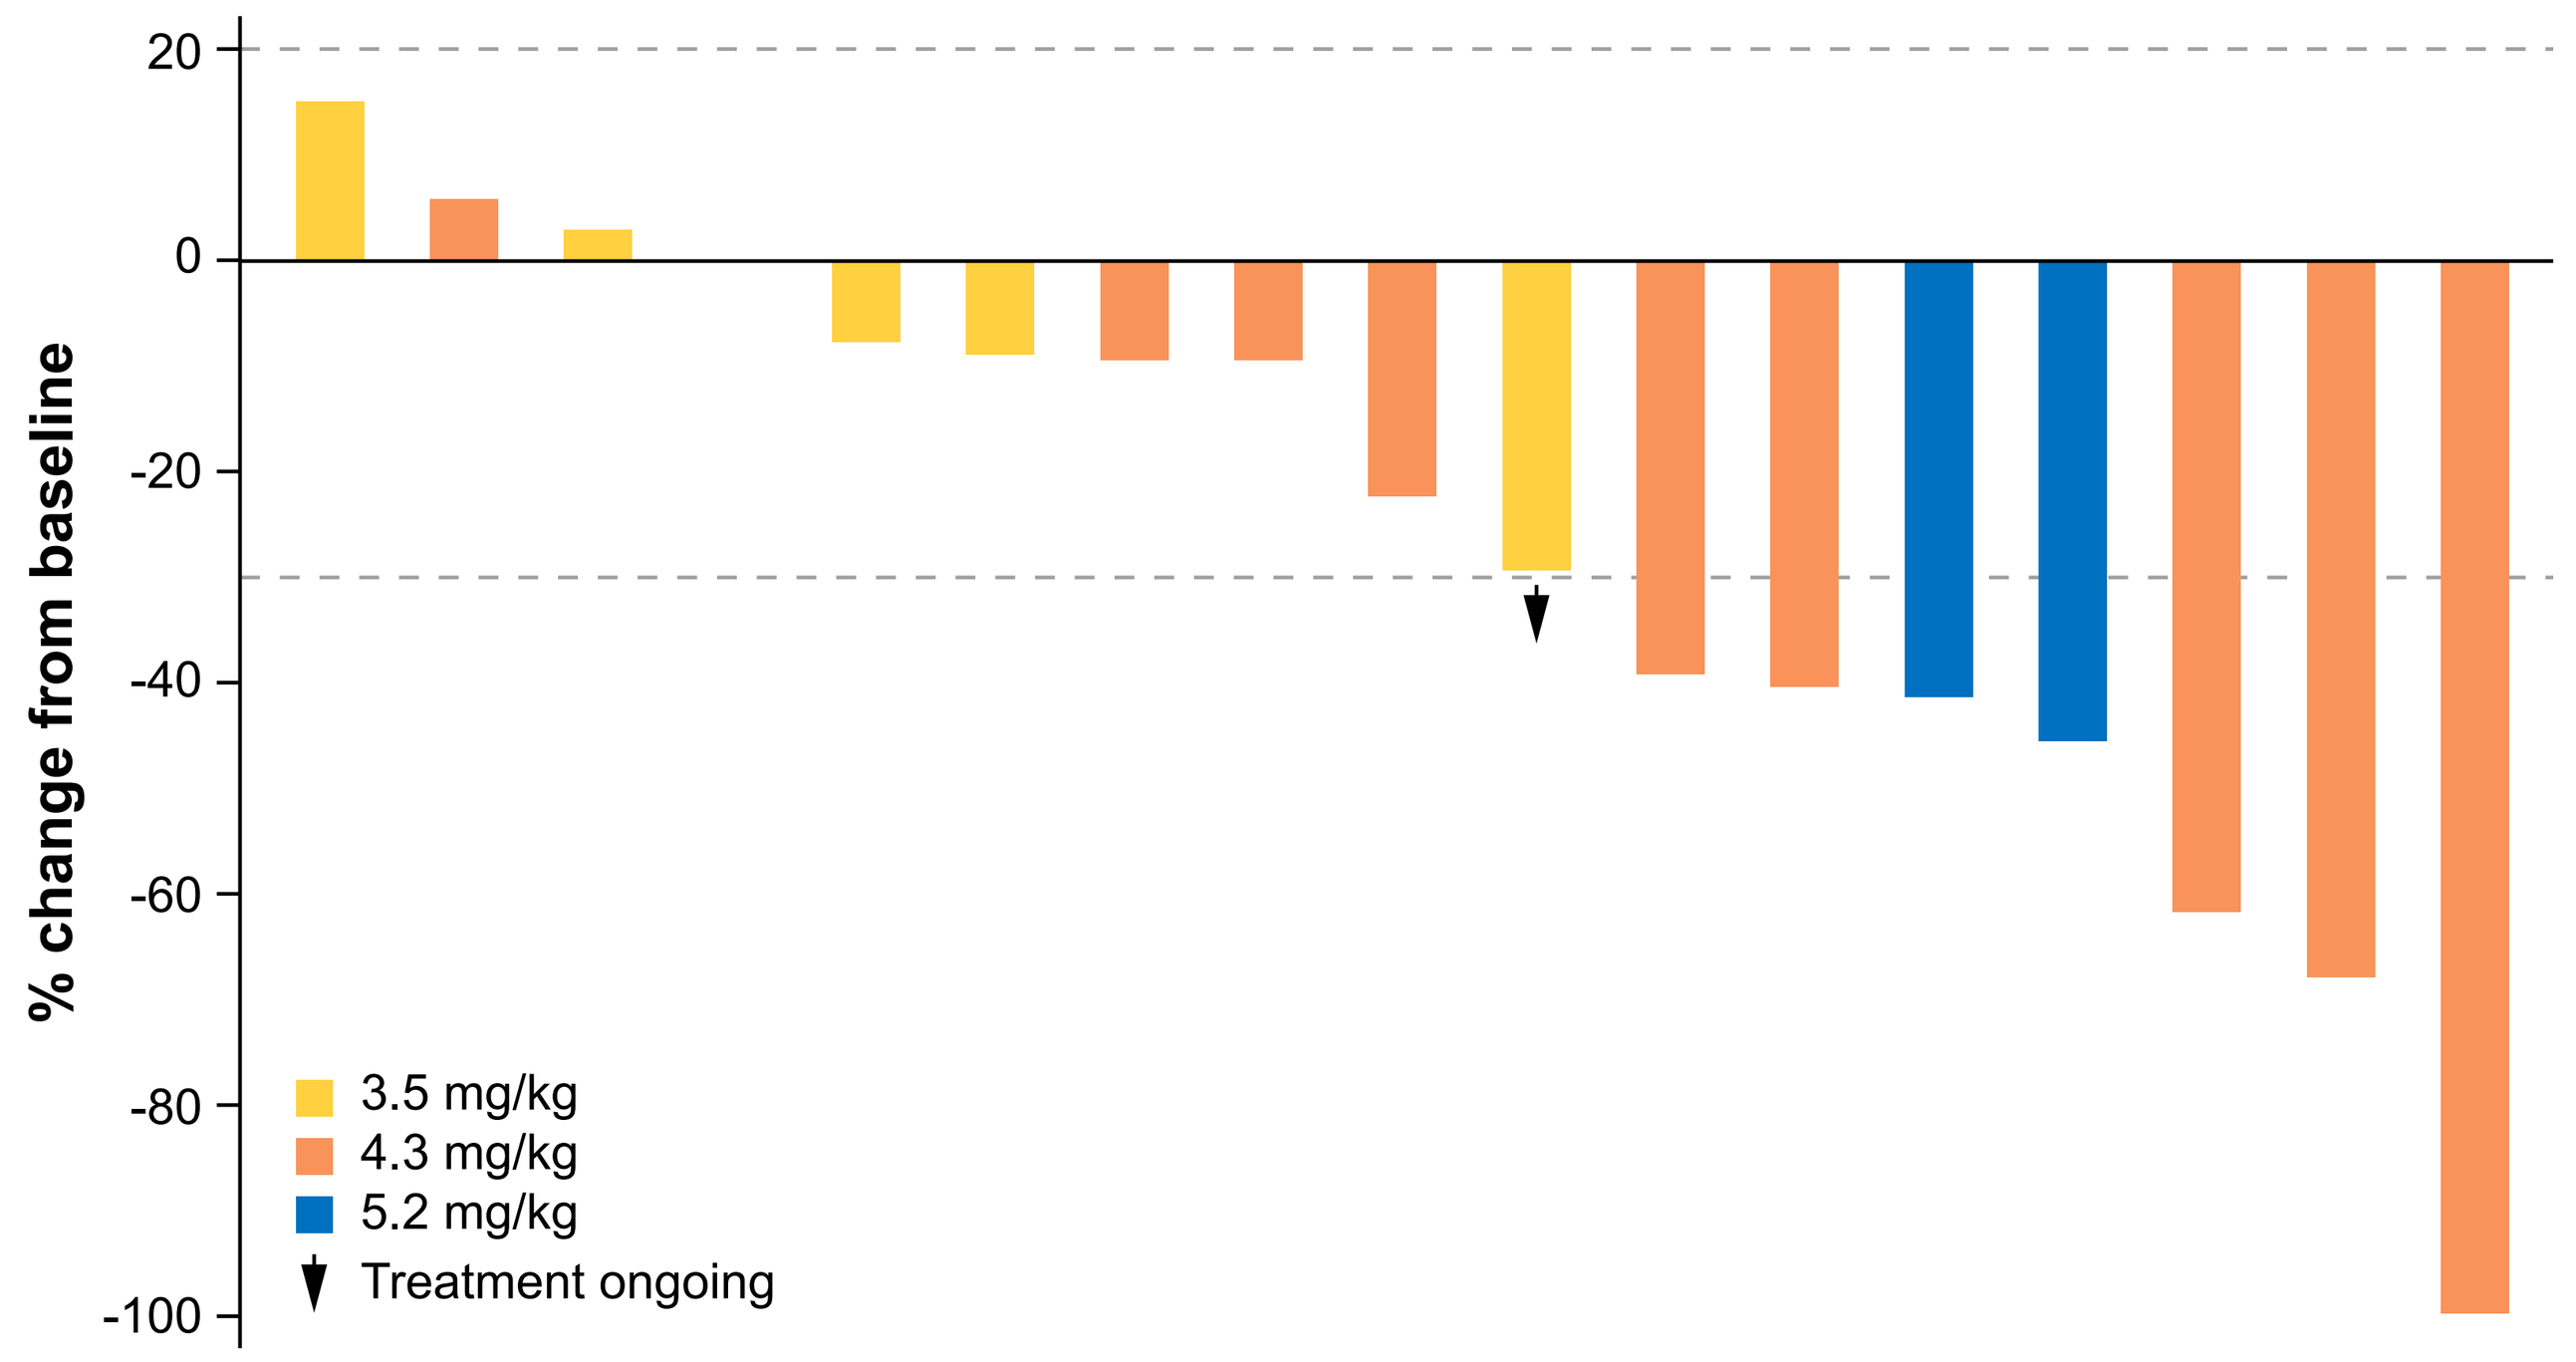
20
0
-20
-40
-60
-80
-100
% change from baseline
3.5 mg/kg
4.3 mg/kg
5.2 mg/kg
Treatment ongoing

Data cut off date: August 2nd, 2024.

CONCLUSIONS

- Luvelta plus bevacizumab demonstrated encouraging antitumor activity irrespective of FRα expression and prior bev treatment
- Luvelta + bev had no new safety signals vs either agent alone
- These early data in combination may offer a non-biomarker driven approach to treat patients with relapsed/recurrent EOC
- The expansion phase of the study is accruing at the selected luvelta dose of 4.3 mg/kg in combination with bev; 23 patients have been enrolled to date

References

1. Cheung A, et al. *Oncotarget*. 2016;7:52553–74.
2. Marchetti C, et al. *Semin Cancer Biol*. 2021;77:144–66.
3. Li X, et al. *Mol Cancer Ther*. 2023;22:155–67.
4. Oaknin A, et al. *J Clin Oncol*. 2023;41(16 suppl):5508.
5. O'Malley DM, et al. *Gynecol Oncol*. 2020; 157:379–85.

Acknowledgements

Editorial and medical writing support was provided by Sandra Mendes, PhD, CMPP, from Aptitude Health, The Hague, the Netherlands, and funded by Sutro Biopharma.

Disclosures

Sutro Biopharma funded this study and participated in the study design, research, data collection and analysis and review and approval of this poster.
A. Madariaga: Astrazeneca (Invited Speaker, Advisory Board), Clovis (Invited Speaker), GSK (Invited Speaker, Advisory Board), MSD (Invited Speaker), Pharmamar (Advisory Board). Non-Financial Interests: EORTC, Member, Chair Young Investigators Gynecology Cancer Group.

Presented at the European Society for Medical Oncology (ESMO) Congress, 13–17 September 2024, Barcelona, Spain, and Online

Presenting author contact: ainhoama@hotmail.com